Exclusion of CD8 T Cells from Tumor Epithelium is Associated with Worse Immune Checkpoint Blockade (ICB) Therapy Response and Survival in Non-Small Cell Lung Cancer (NSCLC)

Xinwei Sher[1], Fredrick Gootkind[1], Yanchao Wang[1], Jean-Philippe Guégan[2], Alban Bessede[2], Italiano Antoine[3], G. Travis Clifton[1], Laura A. Dillon[1]

[1] Incendia Therapeutics, Cambridge, MA, USA; [2]Explicyte Immuno-Oncology, Bordeaux, France; [3]Early Phase Trials and Sarcoma Unit, Institut Bergonié, Bordeaux, France, and University of Bordeaux, Bordeaux, France

INCENDIA THERAPEUTICS

Bergonié UNICANCER NOUVELLE-AQUITAINE

#574

Background

Tumor-infiltrating lymphocytes (TILs) are crucial for patient response to ICB treatment and prognosis [1]. However, tumors characterized by immune exclusion, where T cells are concentrated in the tumor-associated stroma but sparsely present or absent in the tumor epithelium, may not respond well to ICB therapy [2]. Discoidin Domain Receptor 1 (DDR1), a tumor expressing collagen-binding receptor tyrosine kinase, has been implicated in cancer invasion, progression, and immune exclusion [3]. This study aims to utilize multiplex immunofluorescence (mIF) staining of NSCLC tumors to elucidate the impact of immune exclusion on ICB response and prognosis, and to evaluate TIL location in the tumor microenvironment (TME) and associated gene signatures as potential biomarkers of response, and DDR1 as a potential therapeutic target.

Methods

Tumors from 100 NSCLC patients enrolled in the Institut Bergonié profiling precision medicine study (NCT02534649) were evaluated by RNA-seq and mIF staining with a panel containing PanCK, COL1A1, CD8, and DDR1, followed by image analysis (Fig. 1 A, B). Patients were split evenly between Lung Adenocarcinoma (LUAD) and Lung Squamous Cell Carcinoma (LUSC), and between responders and non-responders to ICB therapy. A continuous Immune Exclusion Score (IES) was computed based on CD8 T cell densities in the tumor epithelium and tumor stroma (Fig. 1 C). CD8 T cell density (whole tumor, tumor epithelium, and tumor stroma) and IES were compared in responders vs non-responders and evaluated against survival to identify potential biomarkers of ICB response. Gene signatures associated with IES were identified and tested in an independent lung cancer cohort with ICB outcome data [4].

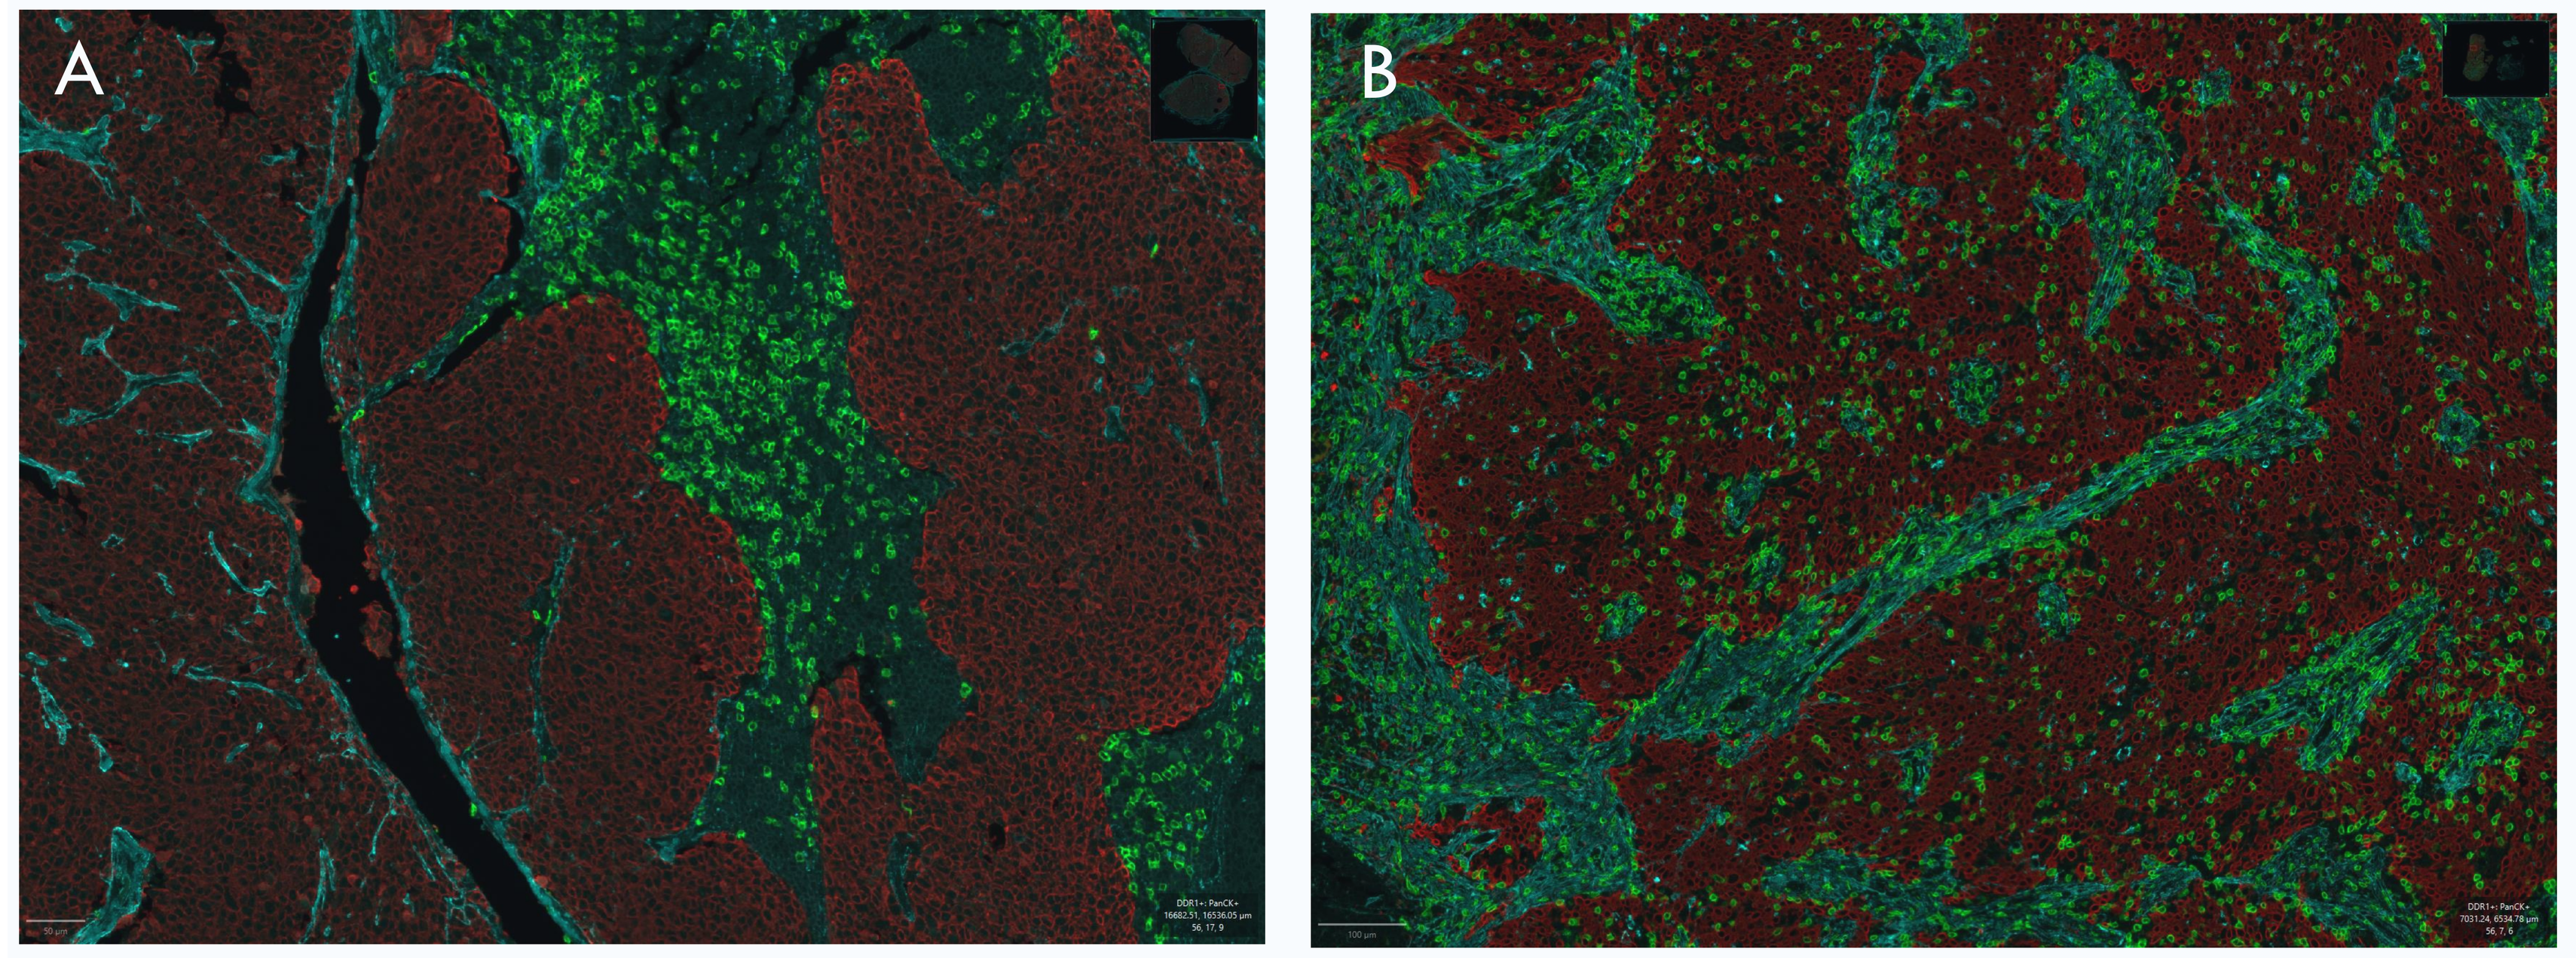
A
B

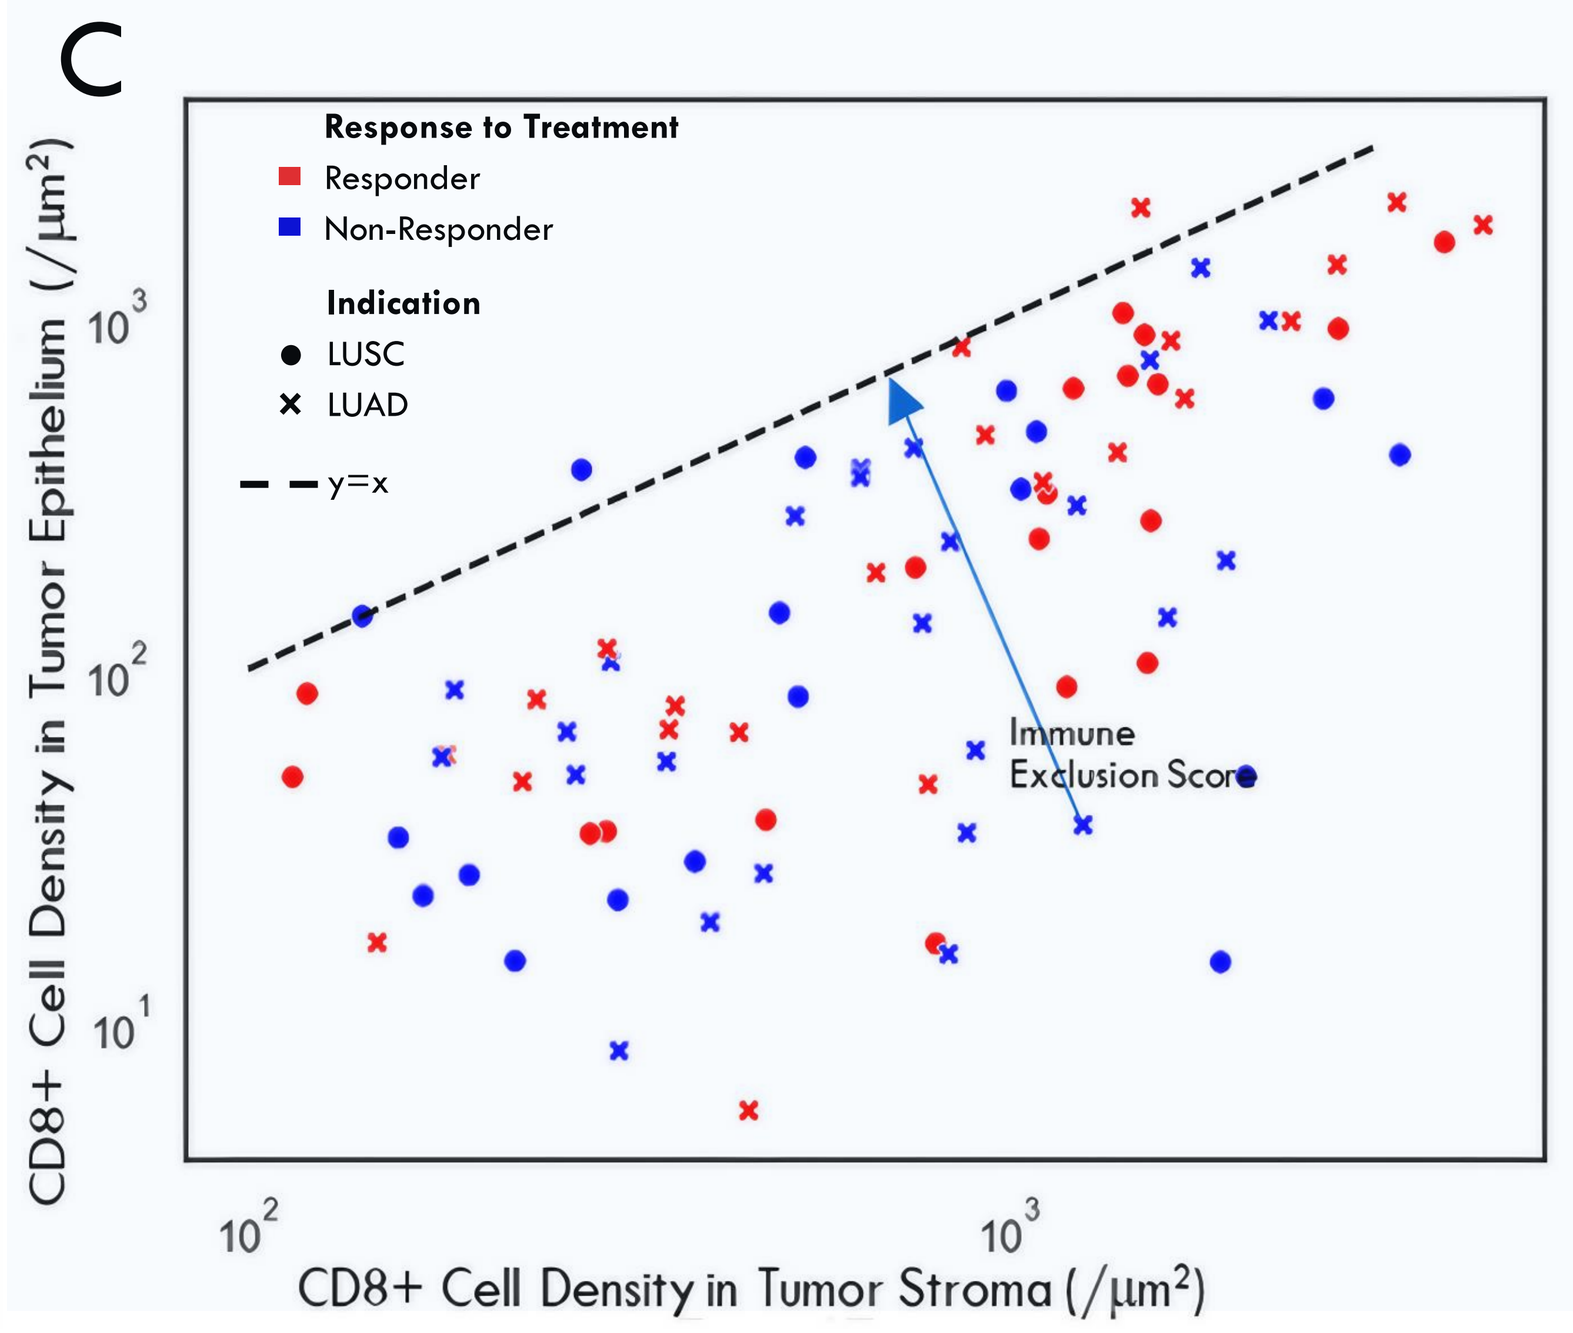
C
Response to Treatment
Responder
Non-Responder
Indication
LUSC
LUAD
y=x
Immune Exclusion Score
CD8+ Cell Density in Tumor Epithelium (/µm²)
CD8+ Cell Density in Tumor Stroma (/µm²)

Figure 1. CD8 T cell (green) distribution in the tumor epithelium (red) and stroma (cyan) revealed distinct tumor immune phenotypes.

A) Immune Exclusion: CD8 T cells enriched in the stroma regions; **B) Immune Infiltration**: CD8 T cells are distributed within the tumor epithelium regions; **C) Immune Exclusion Score (IES)**: distance between tumor's coordinate by CD8 T cell density in stroma and tumor epithelium, and the y=x line, which represents a free distribution of CD8 T cells throughout the tumor.

Results

Higher Tumor Epithelium CD8 T cell Density In Responders to ICB

CD8 T cell density in the tumor epithelium was higher in responders than non-responders, while no difference in CD8 T cell density in tumor stroma was observed.

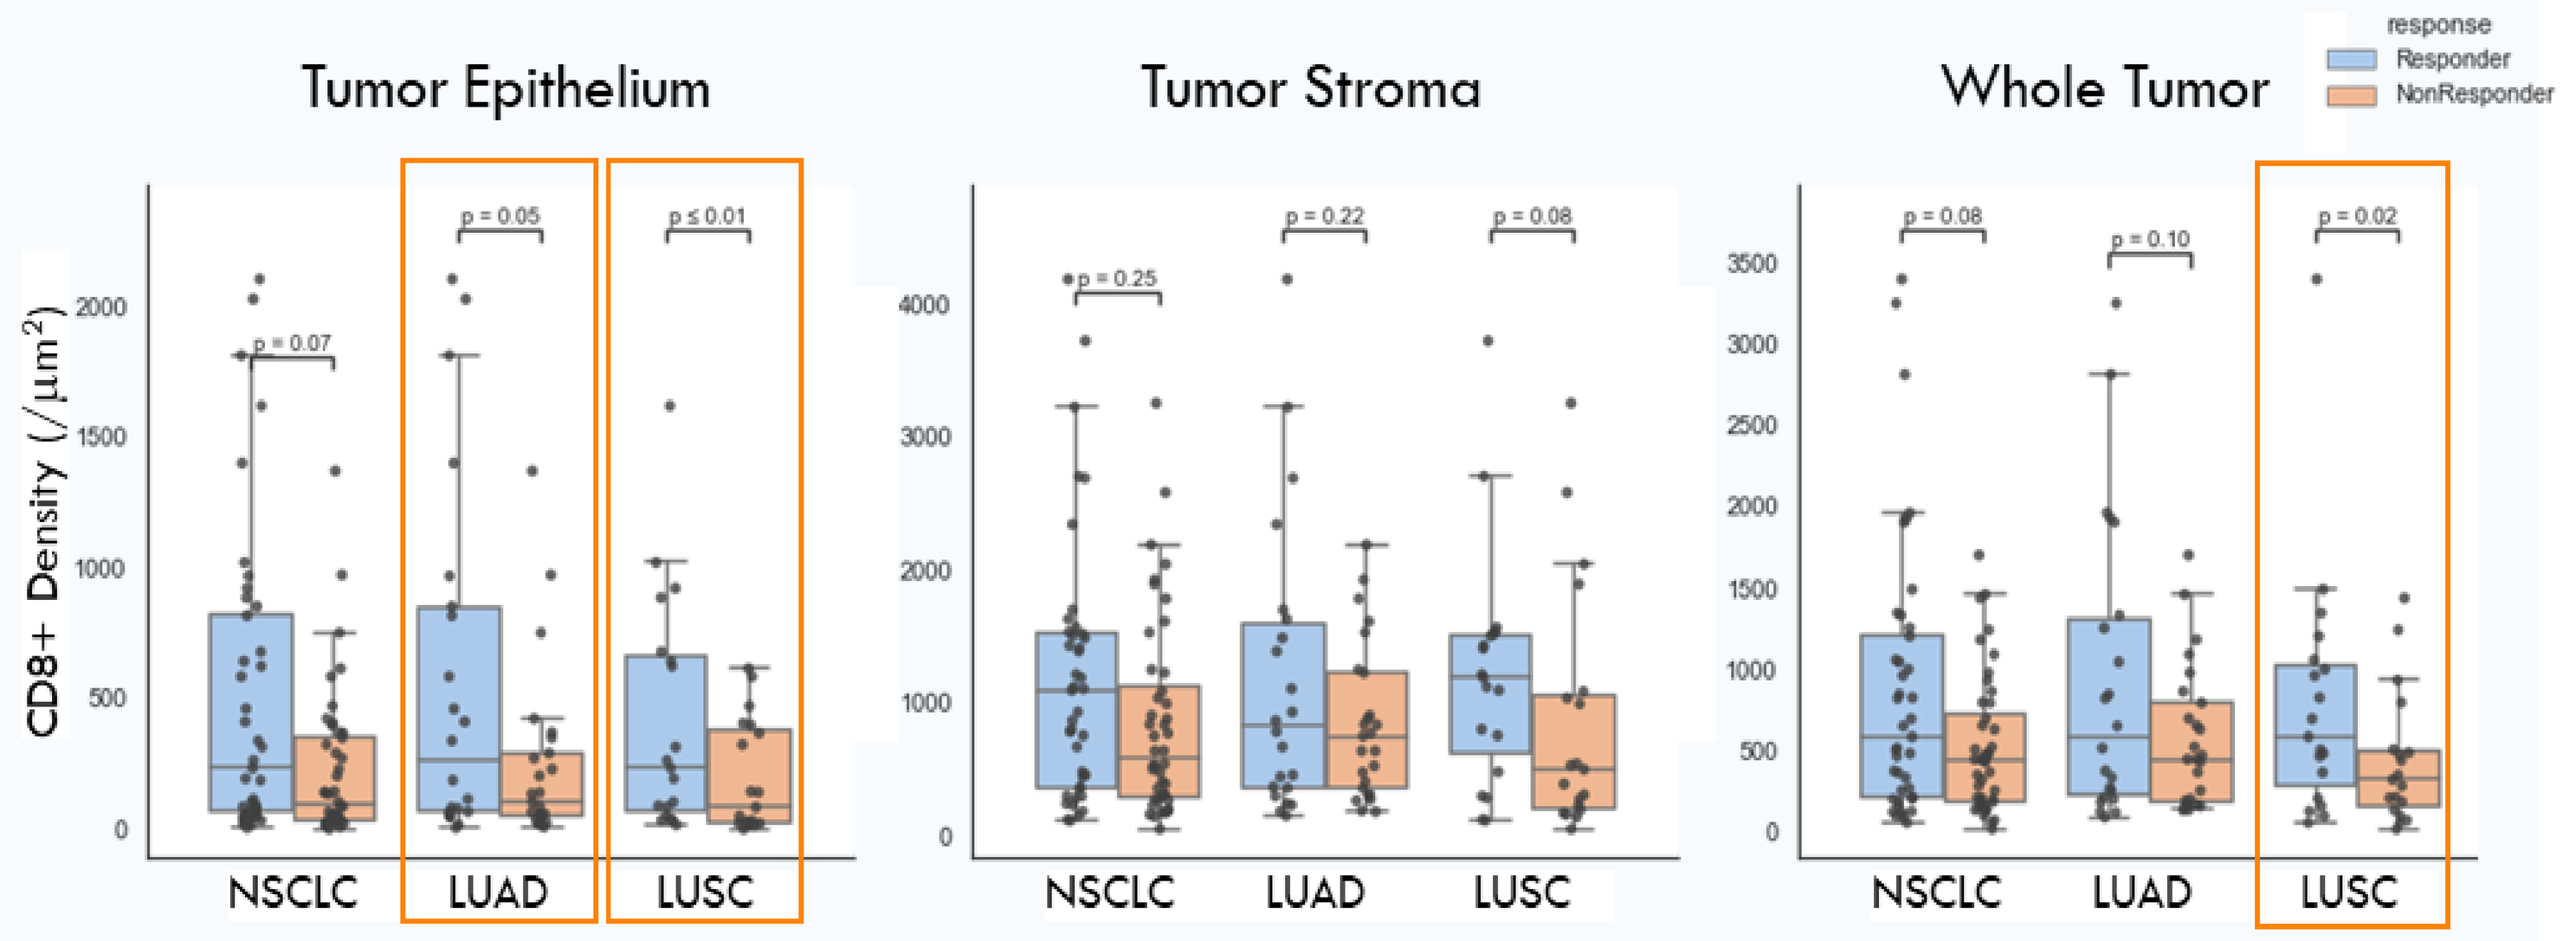
Tumor Epithelium
Tumor Stroma
Whole Tumor
response
Responder
NonResponder
CD8+ Density (/µm²)
NSCLC
LUAD
LUSC

Figure 2. CD8 T cell densities in different tumor regions in NSCLC patients responding or not responding to ICB treatment

This figure compares CD8 T cell densities in the tumor epithelium, stroma, and whole tumor between NSCLC patients who respond and those who do not respond to ICB treatment. The NSCLC cohort includes patients from both LUAD and LUSC subtypes.

CD8 T Cell in Tumor Epithelium, not Stroma, Makes Difference in Patient Survival

Patients with high CD8 T cell density in tumor epithelium, but not in tumor stroma, exhibited improved overall and progression-free survival.

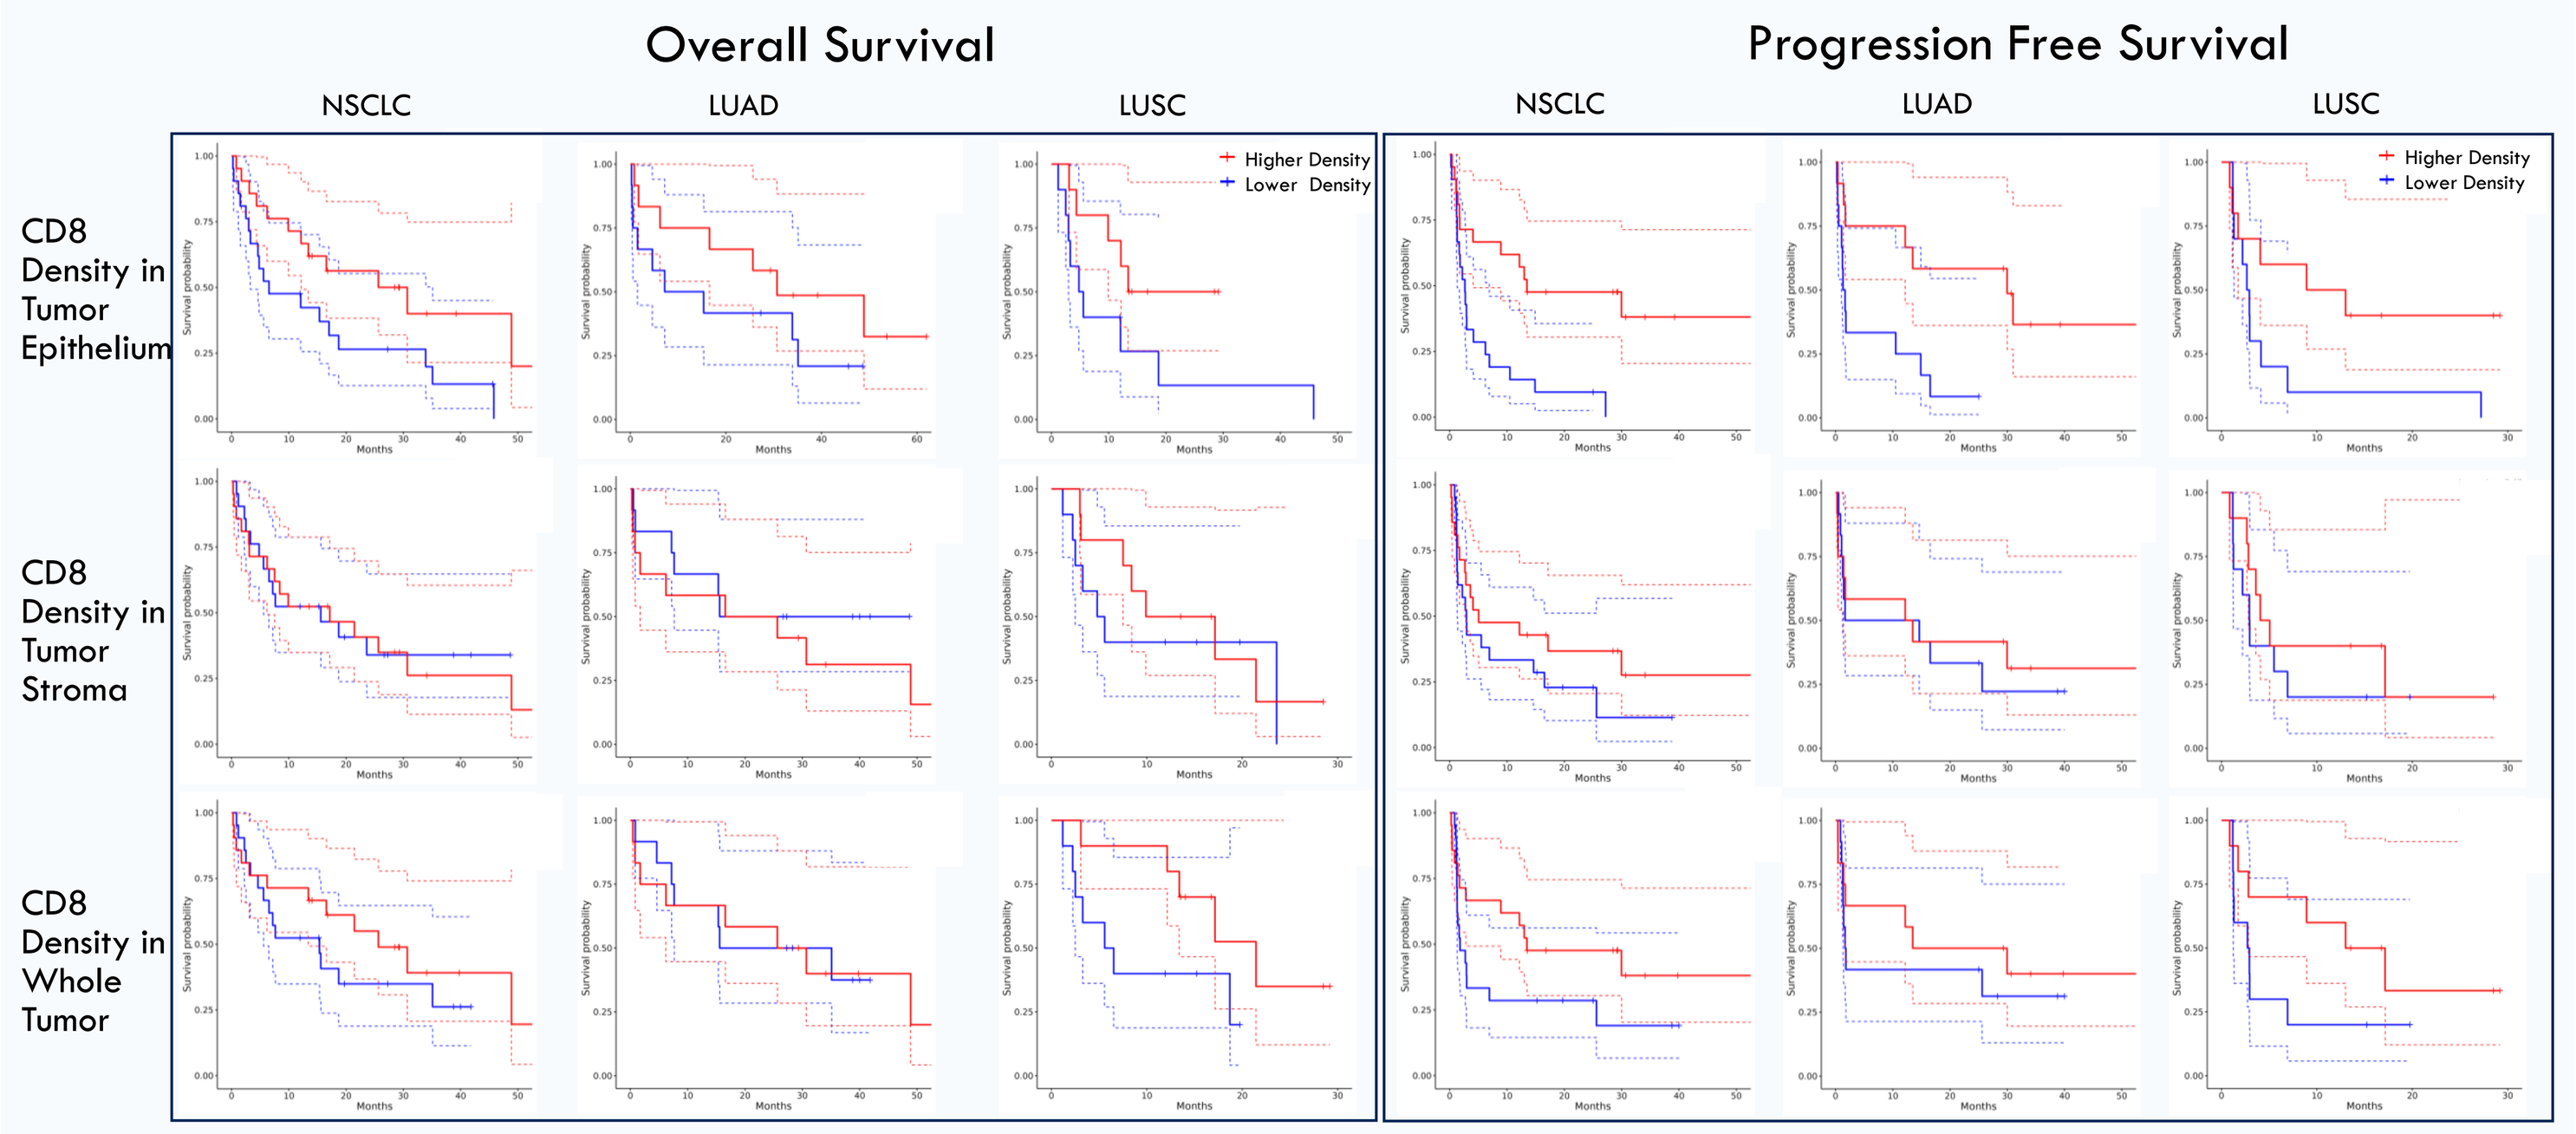
Overall Survival
Progression Free Survival
NSCLC
LUAD
LUSC
Higher Density
Lower Density
CD8 Density in Tumor Epithelium
CD8 Density in Tumor Stroma
CD8 Density in Whole Tumor

Figure 3. Differences in overall survival (OS) and progression-free survival (PFS) in NSCLC patients with high or low CD8 cell densities in tumor epithelium, tumor stroma, and whole tumor

This figure compares OS and PFS between patients with higher CD8 T cell density (top 25%, red) and lower CD8 T cell density (bottom 25%, blue). Higher CD8 T cell density in the tumor epithelium is associated with the greatest survival benefit, while higher CD8 T cell density in the tumor stroma shows very small or no survival benefit. The survival benefit is modest for higher CD8 T cell density in the whole tumor. Dotted lines indicate the 95% confidence intervals.

Immune Exclusion Score (IES) and Patient Overall Survival

Patients with high IES had shorter survival than patients with low IES, independent of PD-L1 expression.

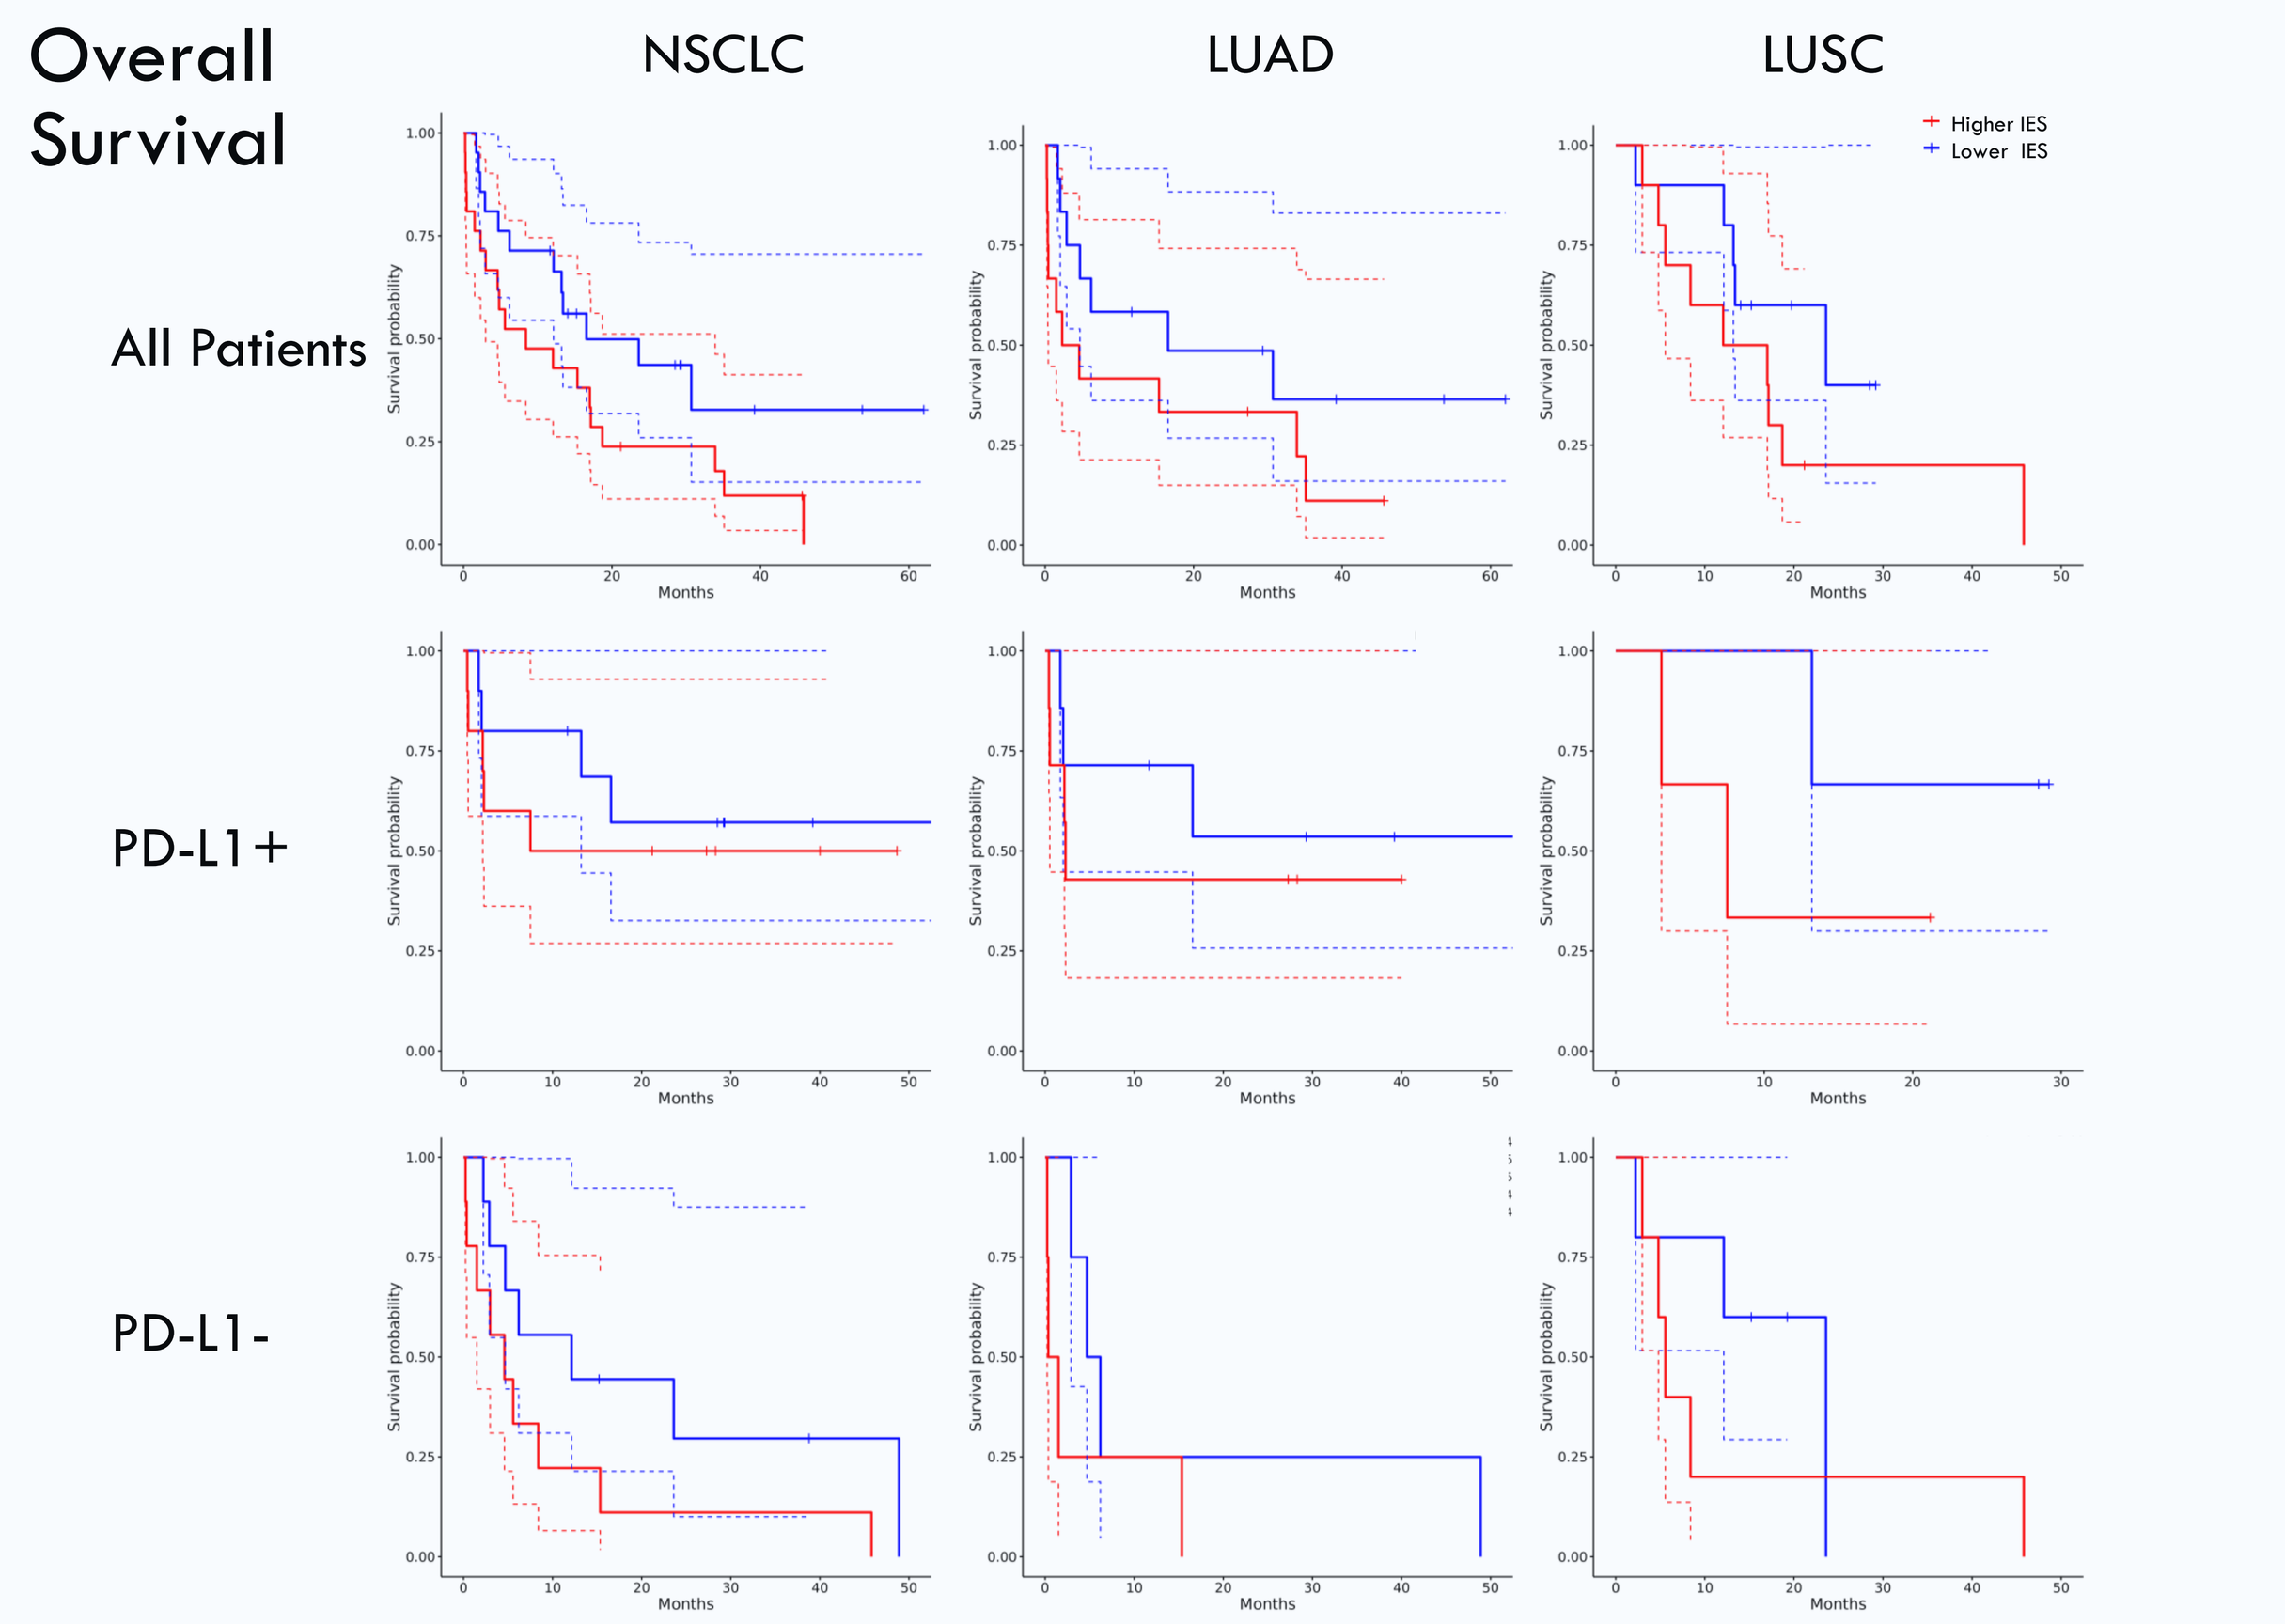
Overall Survival
NSCLC
LUAD
LUSC
Higher IES
Lower IES
All Patients
PD-L1+
PD-L1-
Months

Figure 4. Differences in overall survival (OS) in NSCLC patients with high or low Immune Exclusion Score (IES)

Comparison between higher IES (top 25%, red) and lower IES (bottom 25%, blue) patients. Tumors with high IES exhibit shorter OS and PFS in LUAD, LUSC, and combined cohorts, independent of PD-L1 expression. .Dotted lines indicate the 95% confidence intervals

Independent Validation of Association between CD8 T Cell Exclusion and ICB Response

Expression of the CD8 T cell exclusion signature was higher in ICB non-responders from an independent lung cancer study [4].

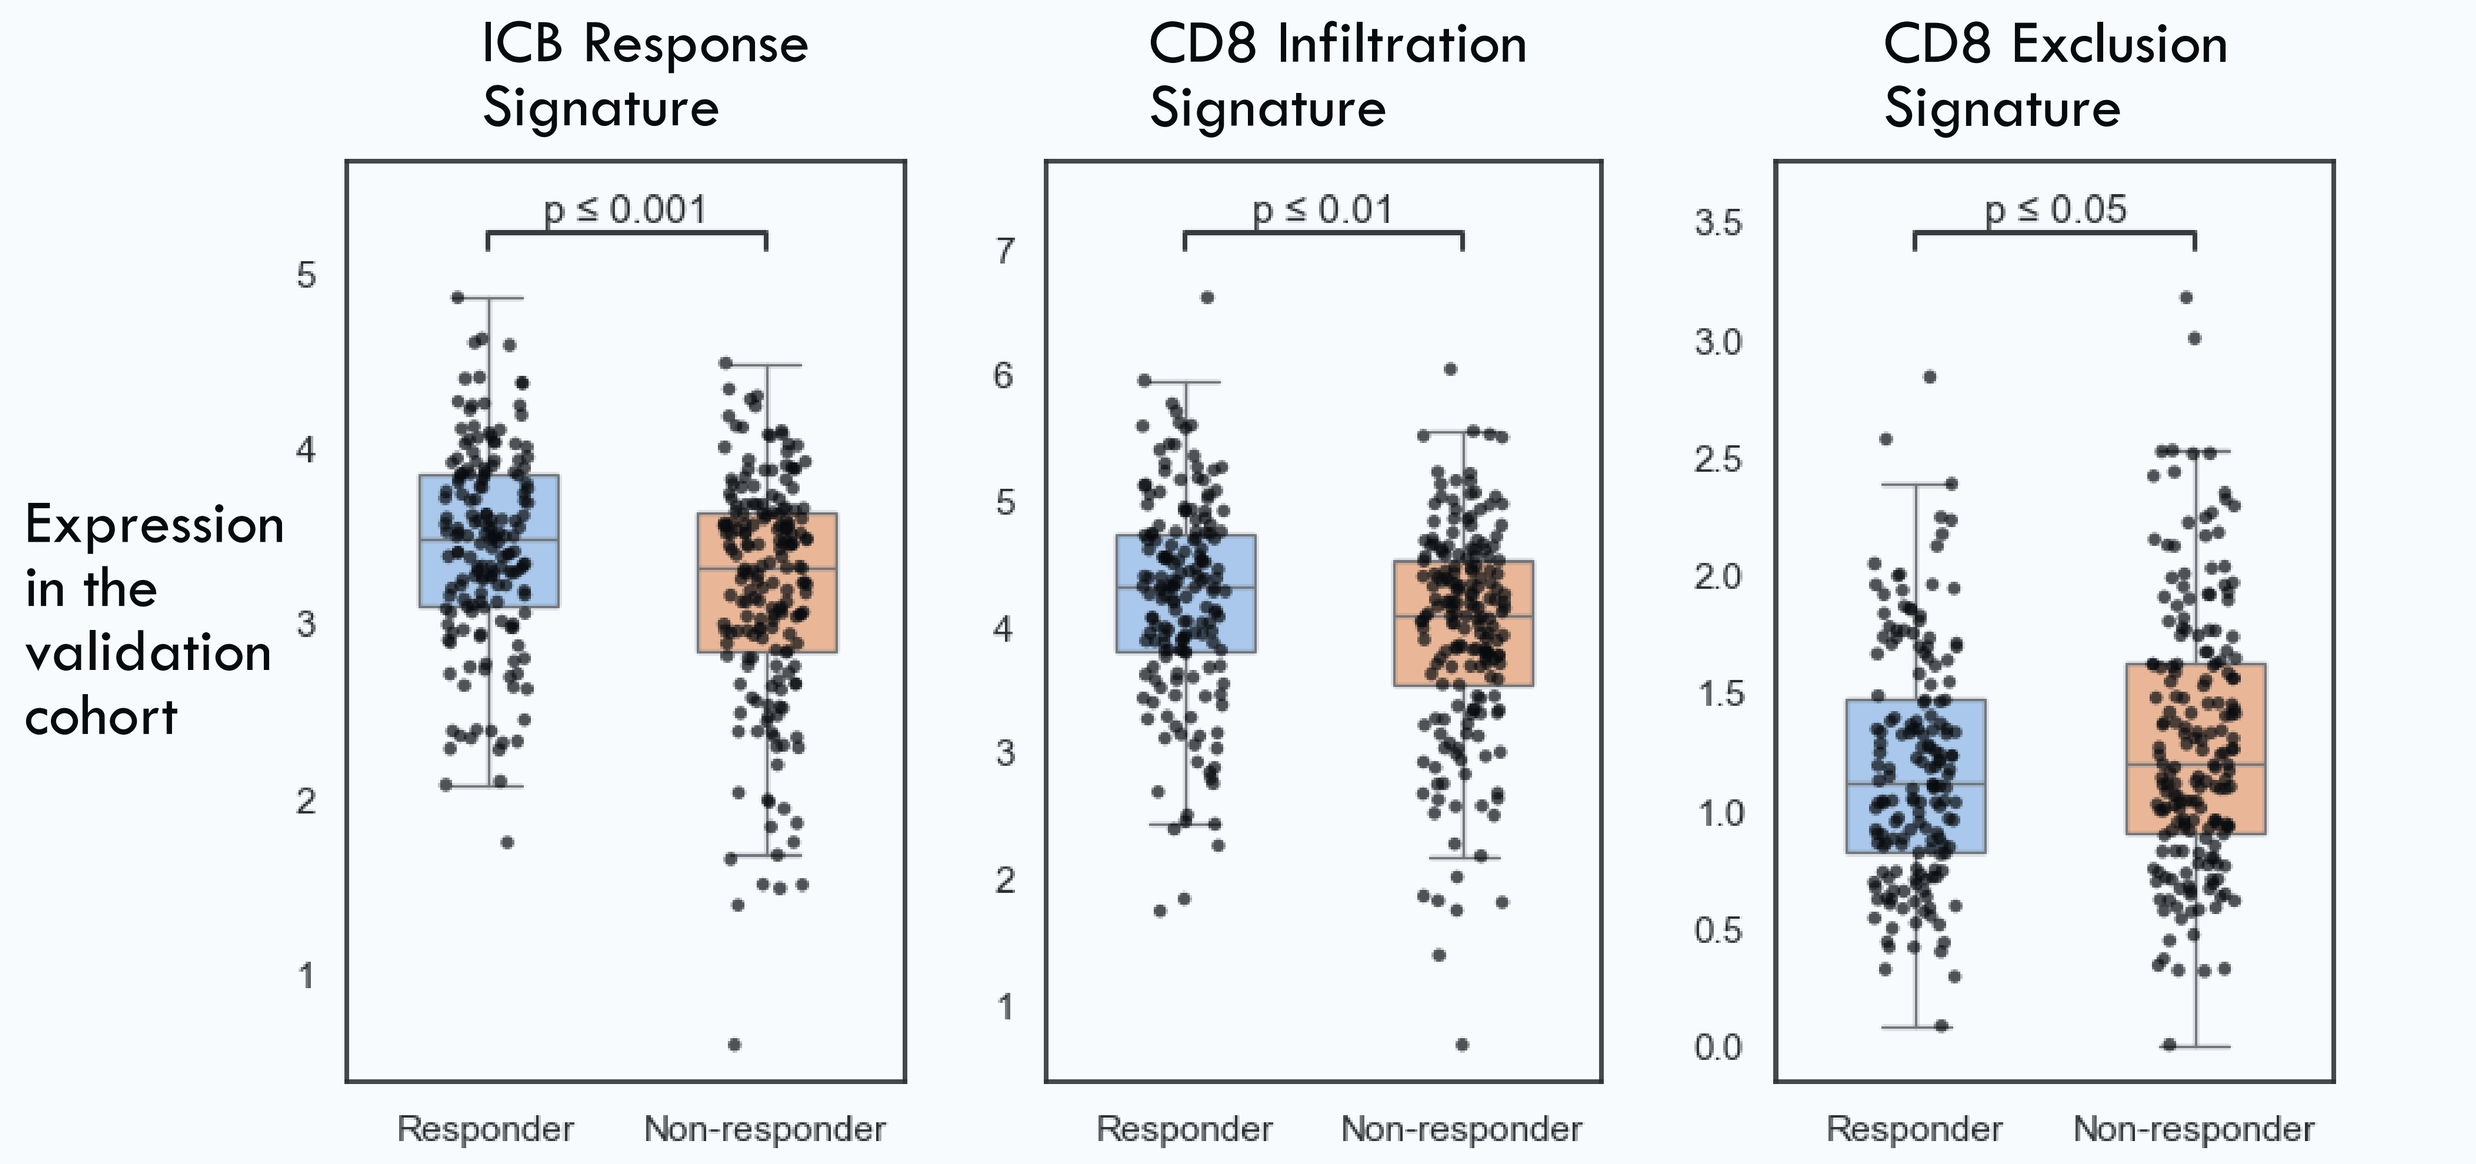
ICB Response Signature
CD8 Infiltration Signature
CD8 Exclusion Signature
p ≤ 0.001
p ≤ 0.01
p ≤ 0.05
Expression in the validation cohort
Responder
Non-responder

Figure 5. Similar trend of expression of ICB response and CD8 infiltration/exclusion gene signatures in an independent cohort

Gene signatures were extracted from the RNA-seq data of our NSCLC cohort. Their expression, when examined in an independent validation cohort of 355 lung cancer ICB-treated clinical samples [4], exhibited a similar trend to that observed in our NSCLC cohort.

Indication Specific Association of Drug Target Gene Expression and Immune Exclusion

DDR1 gene and signature expression were correlated with immune exclusion in LUAD, while DDR1+ cell density (protein expression) was correlated with immune exclusion in LUSC.

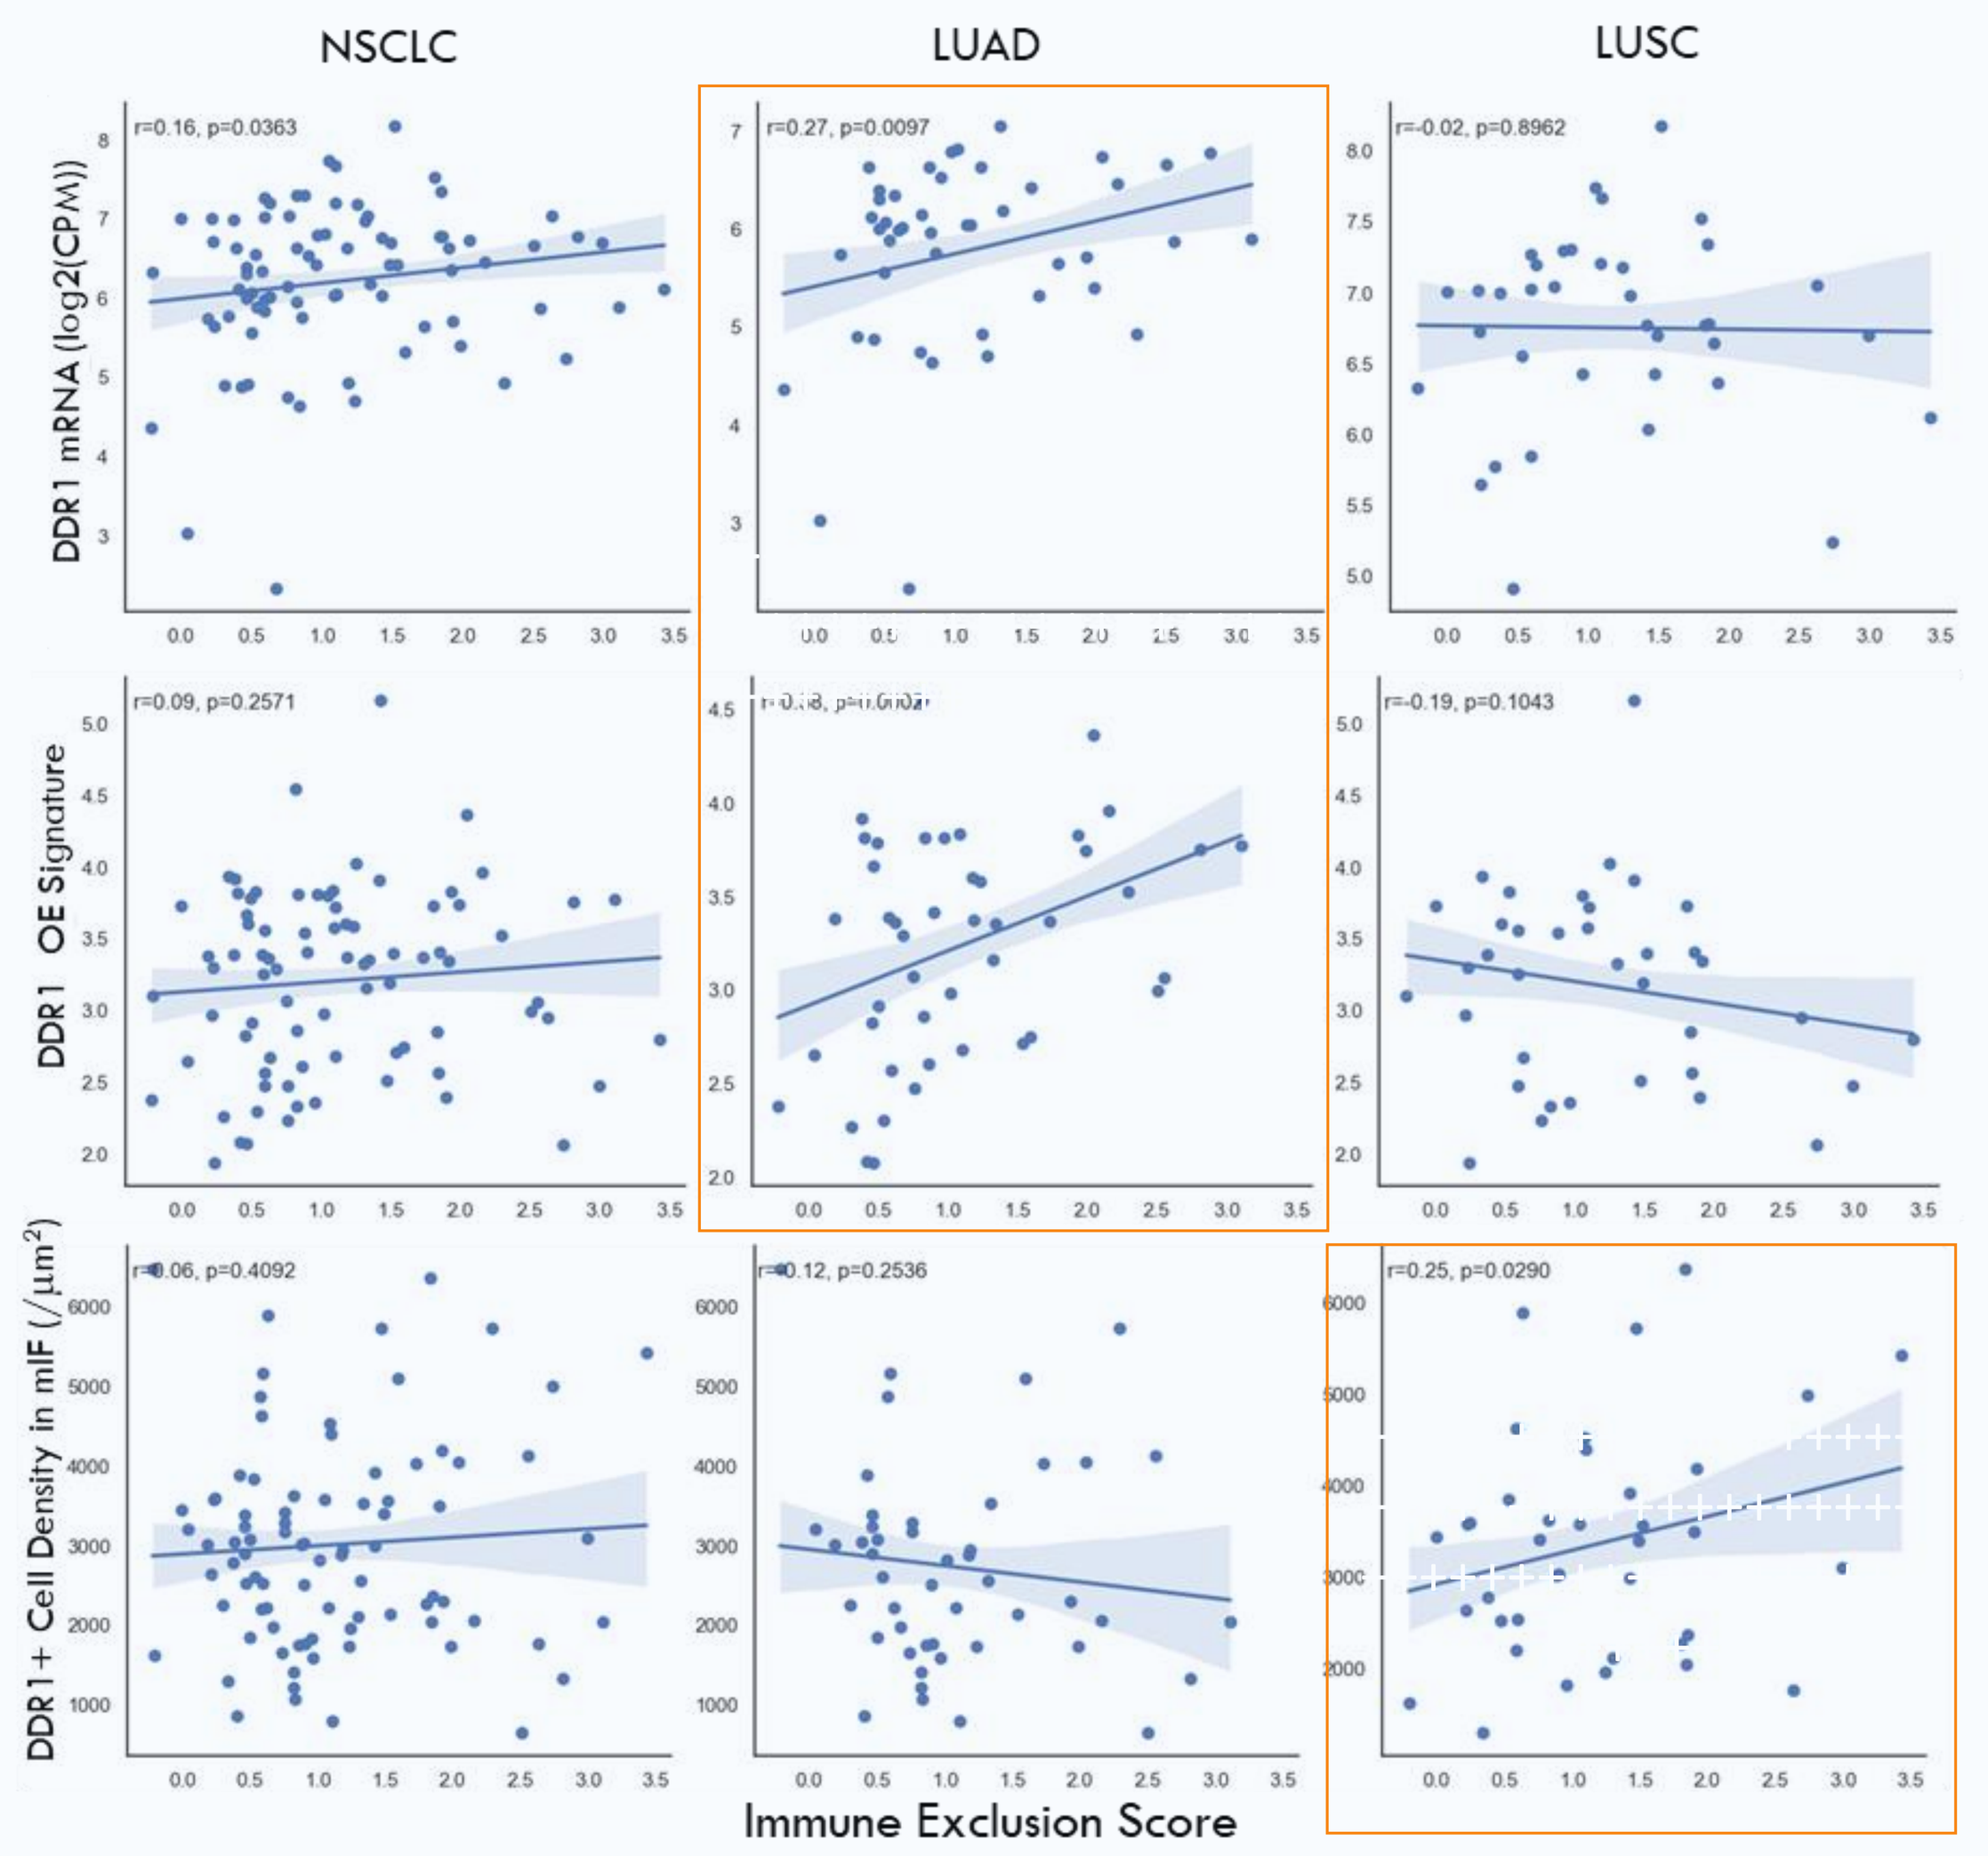
NSCLC
LUAD
LUSC
DDR1 mRNA (log2(CPM))
DDR1 OE Signature
DDR1+ Cell Density in mIF (/µm²)
r=0.16, p=0.0363
r=0.27, p=0.0097
r=-0.02, p=0.8962
r=0.09, p=0.2571
r=-0.19, p=0.1043
r=0.12, p=0.2536
r=0.25, p=0.0290
Immune Exclusion Score

Figure 6. Indication specific DDR1 expression correlation with tumor immune exclusion scores

DDR1 mRNA expression and the DDR1 Overexpression signature (Yang et al., 2022) are strongly correlated with the CD8 T cell-based Immune Exclusion Score in LUAD. DDR1+ cell density in mIF slides is also correlated with the Immune Exclusion Score in LUSC.

Conclusions

CD8 T cell density in the tumor epithelium, rather than the tumor stroma, is associated with response and survival in ICB-treated NSCLC patients. Additionally, CD8 T cell exclusion from the tumor epithelium impedes patient response and reduces survival. The spatial distribution of CD8 T cells within the tumor and immune exclusion-associated gene signatures could serve as potential biomarkers for ICB treatment in NSCLC. DDR1 is a potential therapeutic target to address immune exclusion.

References

1. Loi, S., et al. The journey of tumor-infiltrating lymphocytes as a biomarker in breast cancer: clinical utility in an era of checkpoint inhibition. Ann Oncol. 2021; 32: 1236-1244.
2. Clifton, G.T., et al. Developing a definition of immune exclusion in cancer: results of a modified Delphi workshop. J Immunother Cancer. 2023; 11.
3. Sun, X., et al. Tumour DDR1 promotes collagen fibre alignment to instigate immune exclusion. Nature. 2021; 599: 673-678.
4. Kang, J., et al. Systematic dissection of tumor-normal single-cell ecosystems across a thousand tumors of 30 cancer types. Nat Commun. 2024; 15: 4067.