Abstract ID: 82

PRTH-101, a Discoidin Domain Receptor 1 (DDR1)-Binding Monoclonal Antibody for Treatment of Thymic Cancers

Siraj Sen[1], Funda Meric-Bernstam[2], Alex Spira[3], Aparna Parikh[4], Jordan Berlin[5], David Sommerhalder[6], Patricia LoRusso[7], Rachel E. Sanborn[8], G. Travis Clifton[9], Thomas Schürpf[9], Laura Dillon[9], Susan Macdonald[9], Joseph P. Eder[9], Anthony Tolcher[6]

NEXT Oncology [Dallas, TX (1), Arlington, VA (3), San Antonio, TX (6)], MD Anderson Cancer Center, Houston, TX (2), Massachusetts General Hospital , Boston, MA (4), Vanderbilt-Ingram Cancer Center, Nashville, TN (5), Yale Cancer Center, New Haven, CT (7), Earle A. Chiles Research Institute, Providence Cancer Institute, Portland, OR (8), Incendia Therapeutics, Cambridge, MA (9)

ABSTRACT

Surgery is the primary therapy for thymic cancers, and chemotherapy and immunotherapy for relapsed disease leave considerable room for improvement. DDR1 is a collagen-binding receptor tyrosine kinase expressed by epithelial cells; it is overexpressed on cancer cells and modulates the peritumoral stroma. Thymic epithelial cancers have among the highest levels of DDR1 expression. DDR1 has been implicated in driving cancer invasion, progression, and immune exclusion. DDR1 binding produces highly aligned collagen fibers (Sun *et al*., Nature 2021) that impede effector CD8+ T cell movement into the tumor epithelium, preventing the generation of an inflamed phenotype that is conducive to an effective immune response.

PRTH-101, a monoclonal antibody that binds DDR1, is in a Phase 1 clinical trial as a single agent and together with pembrolizumab. An RP2D has been defined based on pharmacokinetic and pharmacodynamic results, as no dose-limiting toxicities were observed as a single agent or in combination. Forty-five patients have been dosed and 11 remain on trial. Eight patients with thymic cancers have been treated with encouraging early clinical and pharmacodynamic results.

BACKGROUND

DDR1 Expression is Associated with Highly Ordered Collagen and Exclusion of Immune Cells from Tumors; PRTH-101 Blocks Collagen Binding

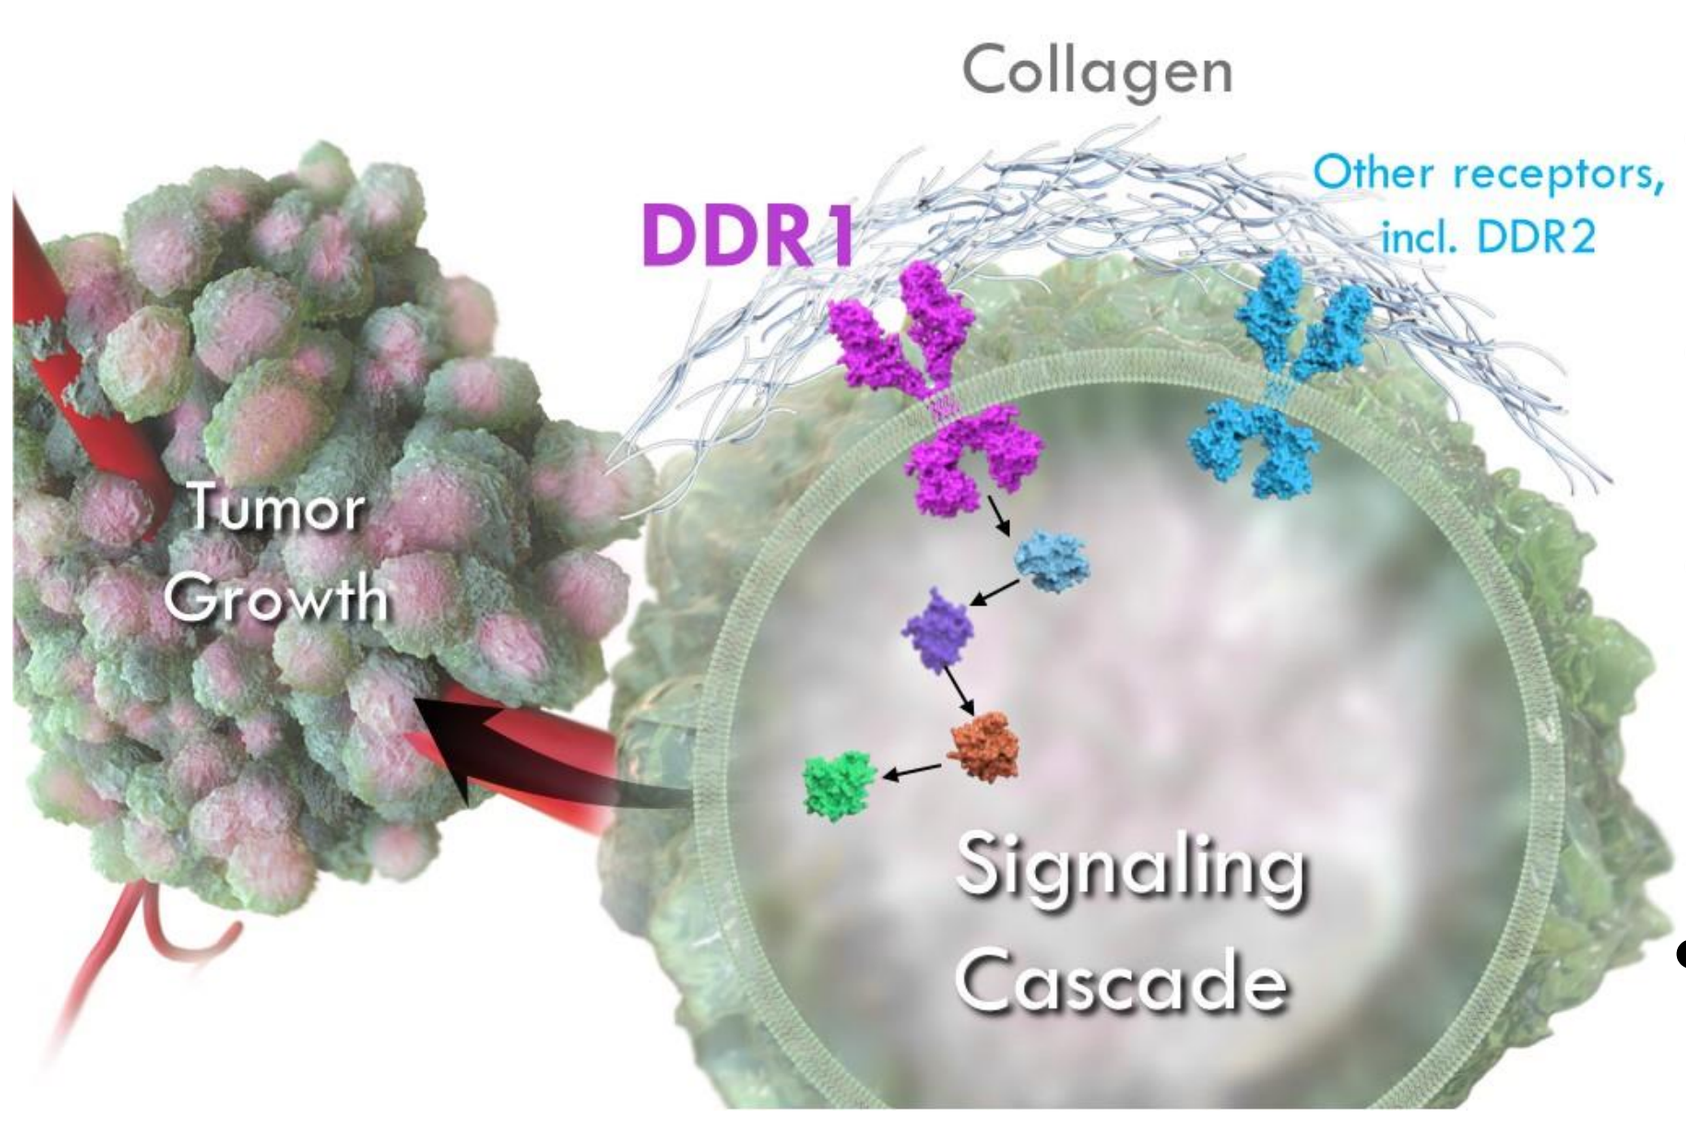
Collagen
DDR1
Other receptors, incl. DDR2
Tumor Growth
Signaling Cascade

- Tumor DDR1 expression is associated with collagen alignment and immune exclusion
- Collagen binding leads to DDR1 receptor clustering, induction of pDDR1, and signaling
- Overexpressed in many cancers and fibrosis, and associated with poor survival
- DDR1 kinase inhibition targeted in cancers
 - Results largely underwhelming
 - Currently no other DDR1 inhibitors in clinical development
- PRTH-101 binds to the extracellular domain of DDR1, blocking collagen binding

Highly Structured Collagens are Prognostic and Predictive in Cancer

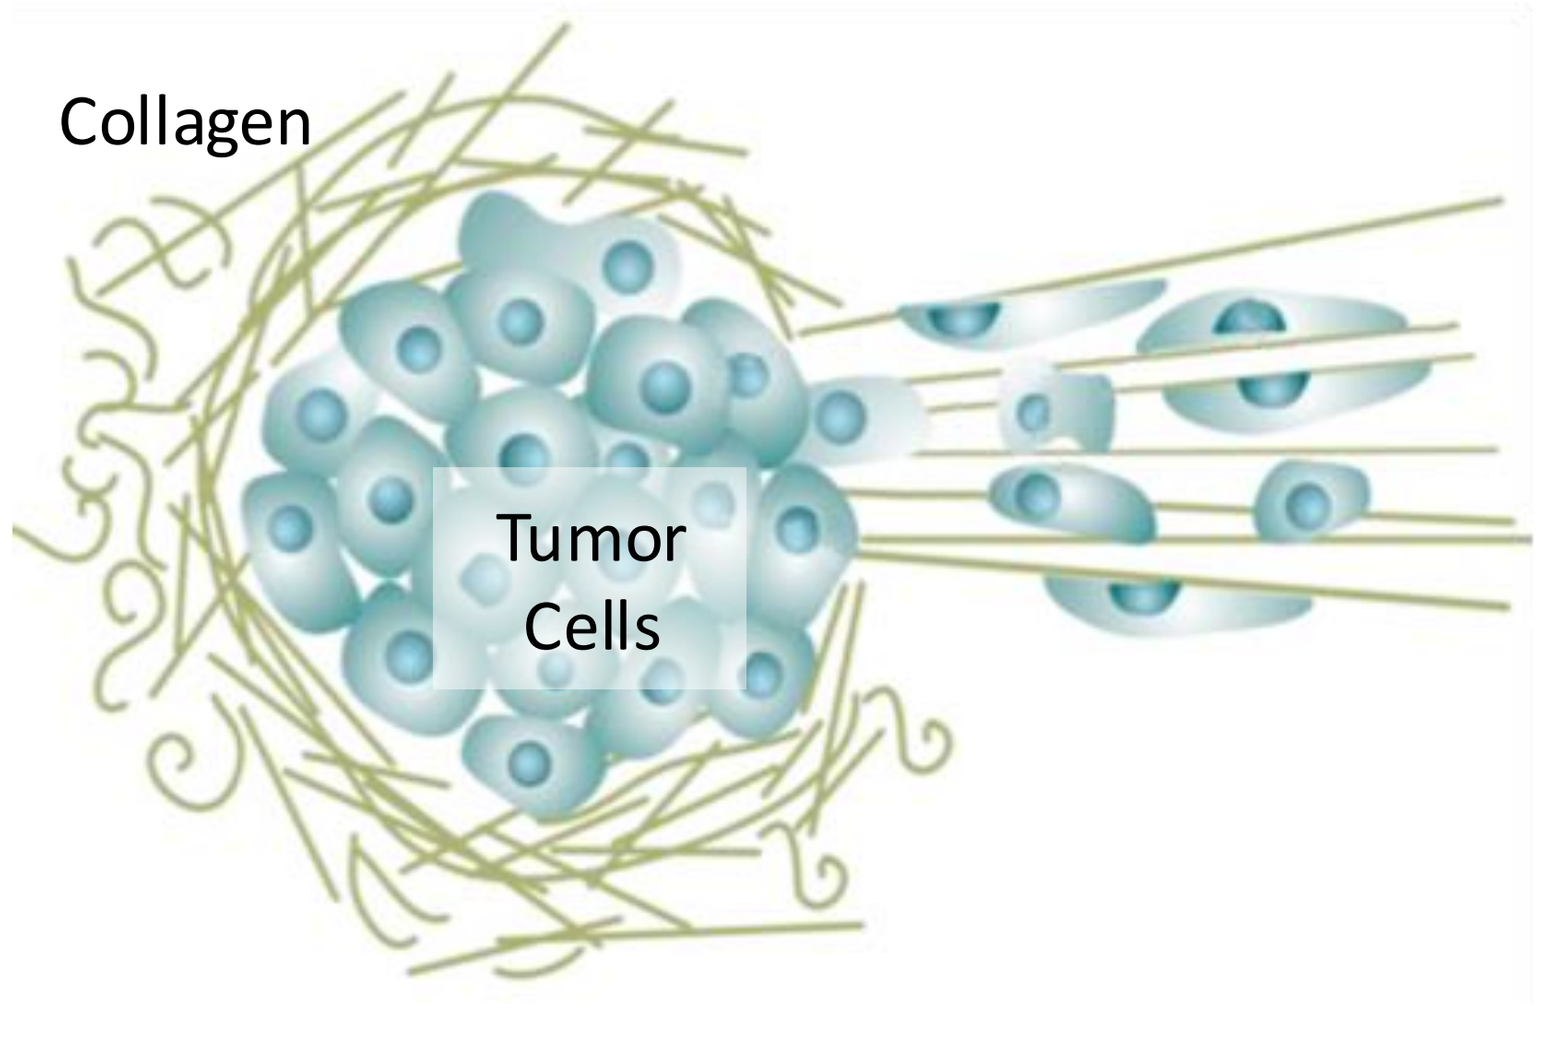
Collagen
Tumor Cells

Tumors use collagen structures to evade the immune system

Adapted from Ray *et al*., Curr. Opin. Cell. Biol., 2021

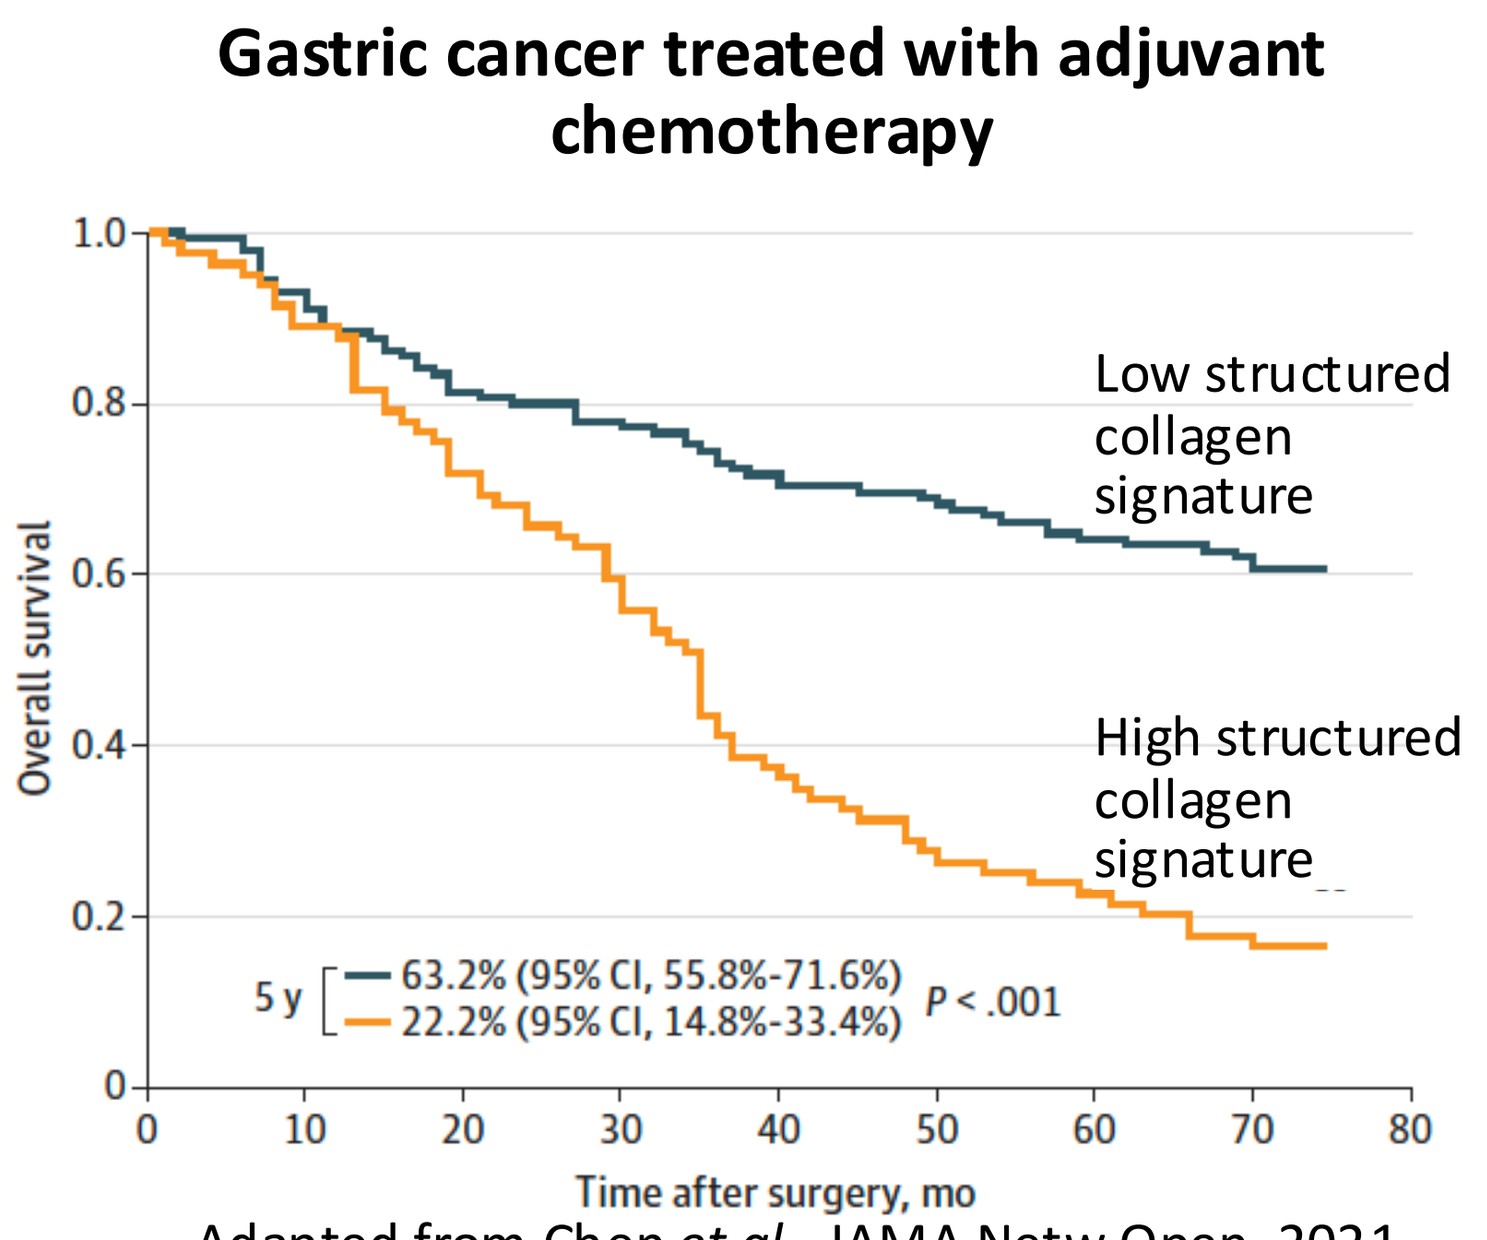
Gastric cancer treated with adjuvant chemotherapy
Overall survival
Low structured collagen signature
High structured collagen signature
5 y
63.2% (95% CI, 55.8%-71.6%)
22.2% (95% CI, 14.8%-33.4%)
P < .001
Time after surgery, mo

Adapted from Chen *et al*., JAMA Netw Open, 2021

High DDR1 Expression is Observed in Multiple Tumor Types

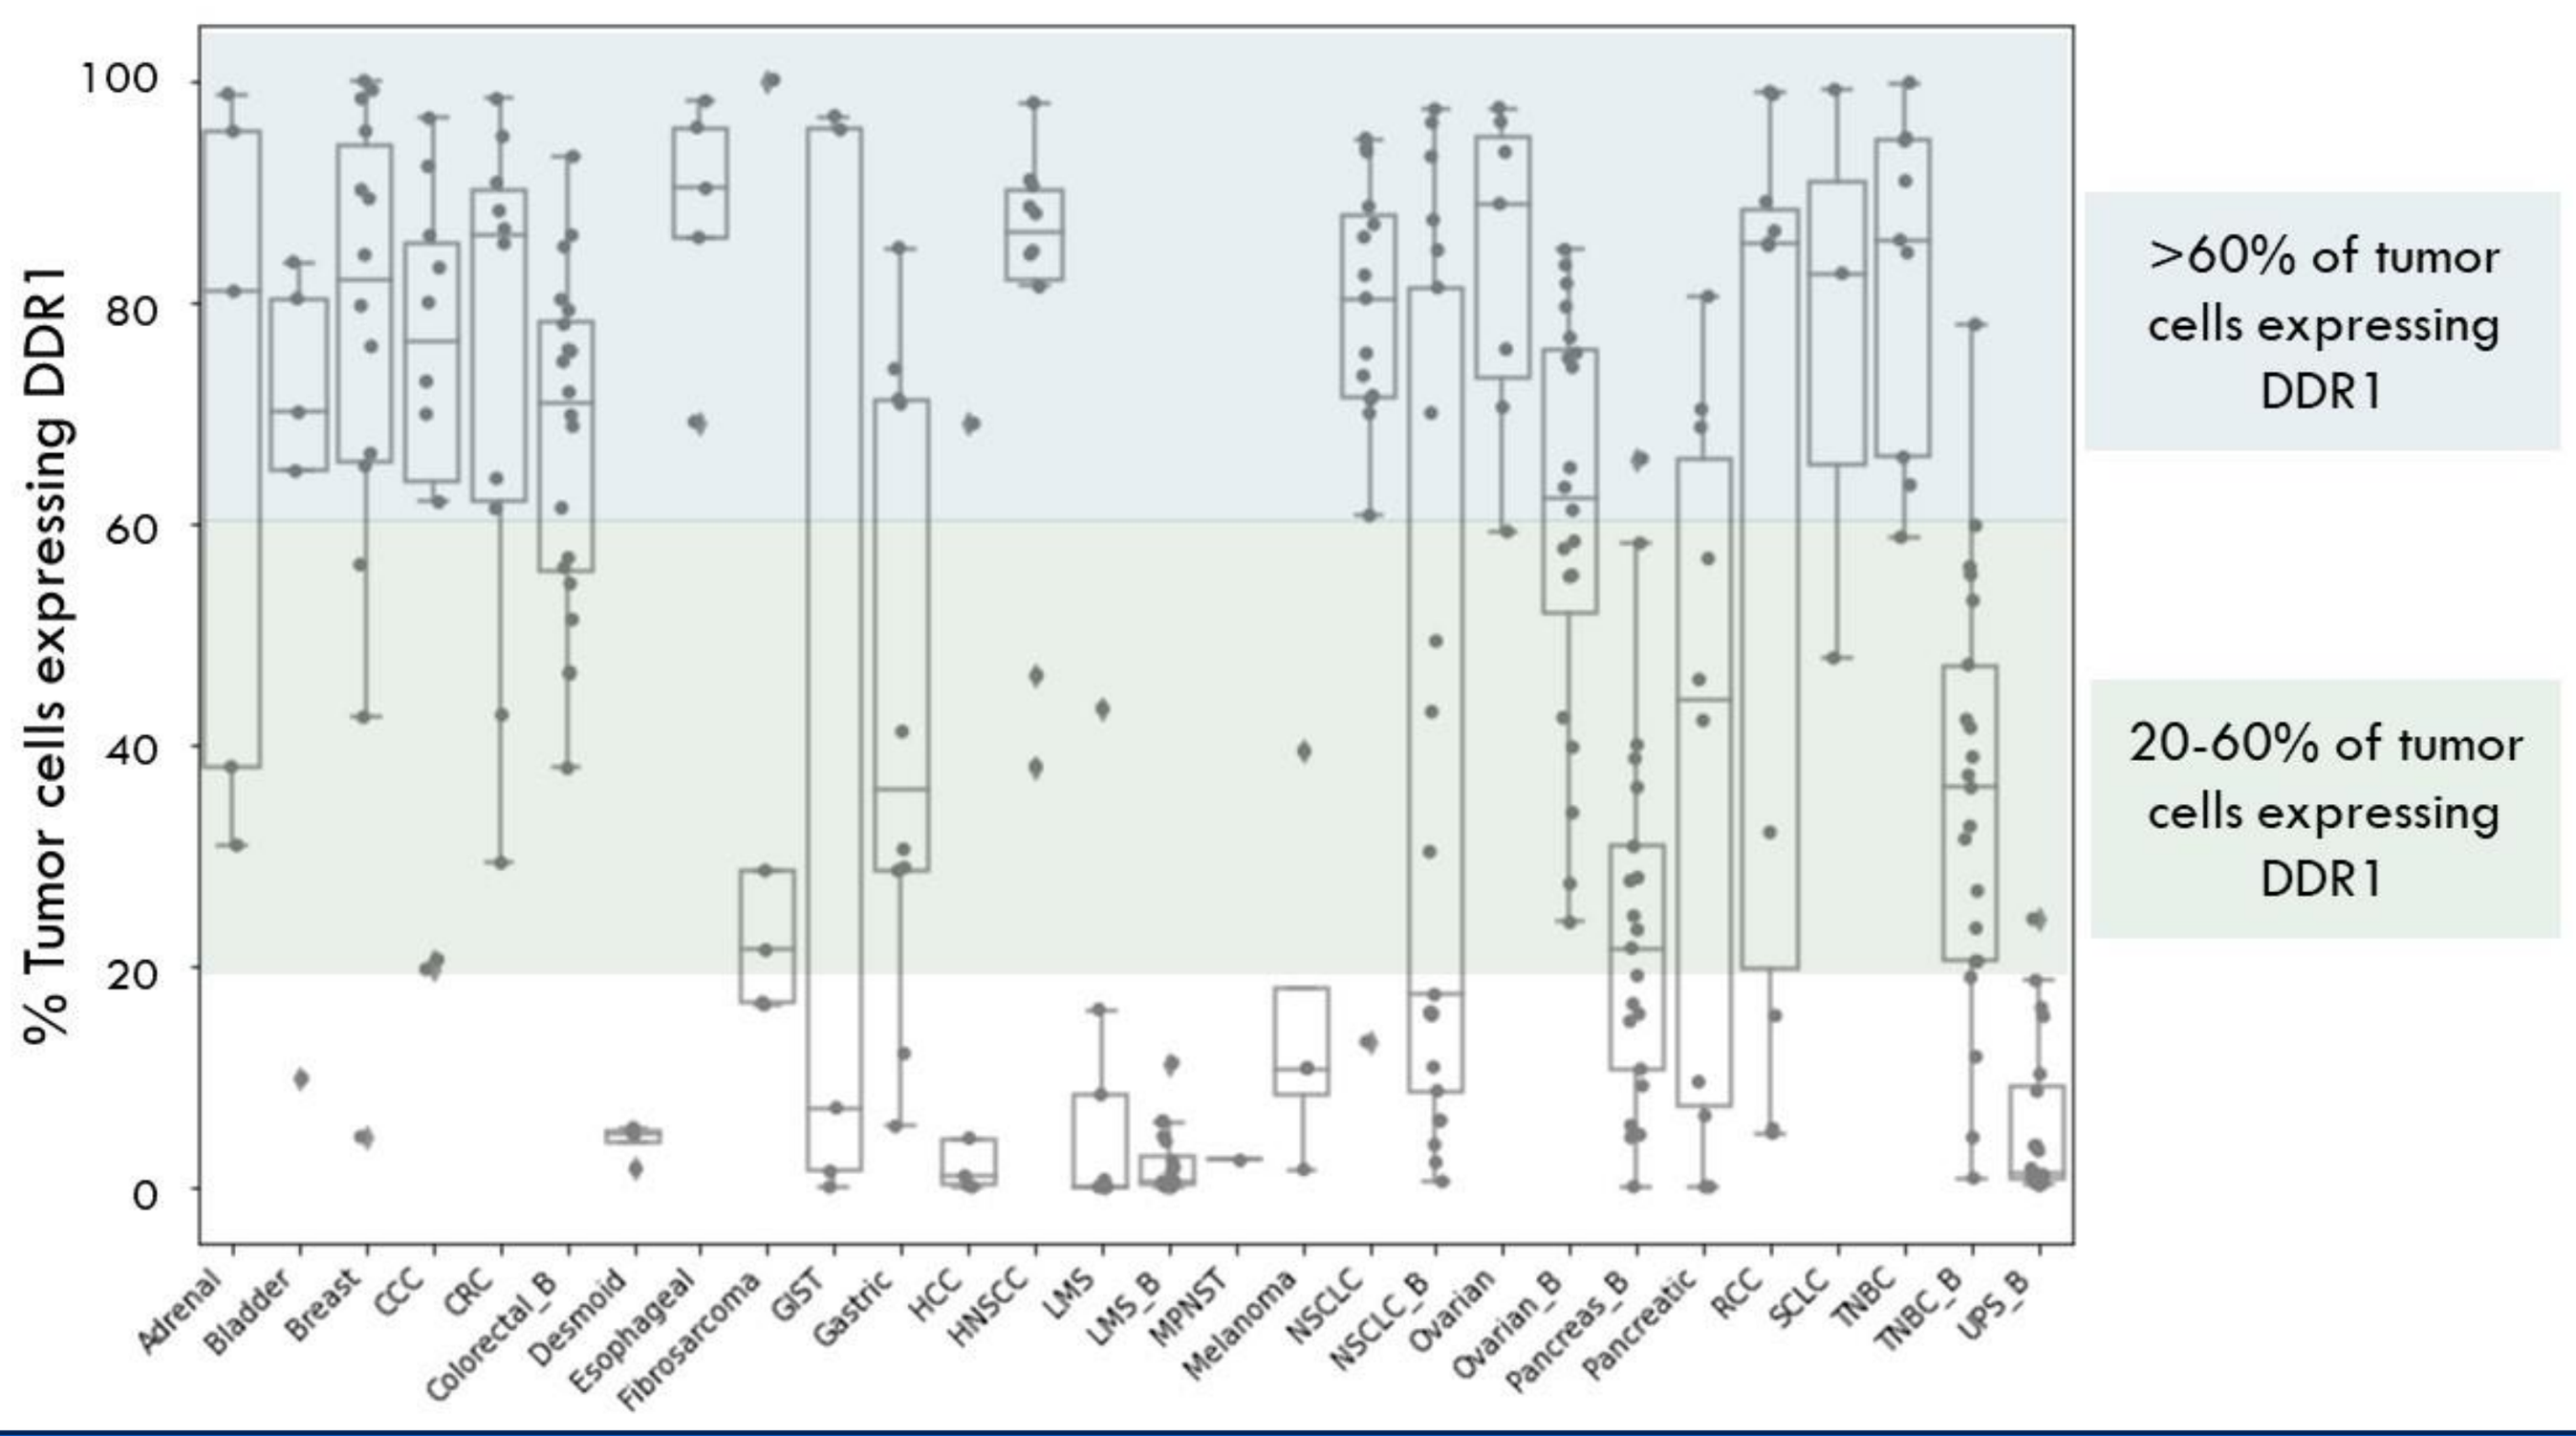
% Tumor cells expressing DDR1
>60% of tumor cells expressing DDR1
20-60% of tumor cells expressing DDR1
Adrenal
Bladder
Breast
CCC
CRC
Colorectal_B
Desmoid
Esophageal
Fibrosarcoma
GIST
Gastric
HCC
HNSCC
LMS
LMS_B
MPNST
Melanoma
NSCLC
NSCLC_B
Ovarian
Ovarian_B
Pancreas_B
Pancreatic
RCC
SCLC
TNBC
TNBC_B
UPS_B

DDR1 is Associated with Resistance to PD-L1 Inhibitor Therapy

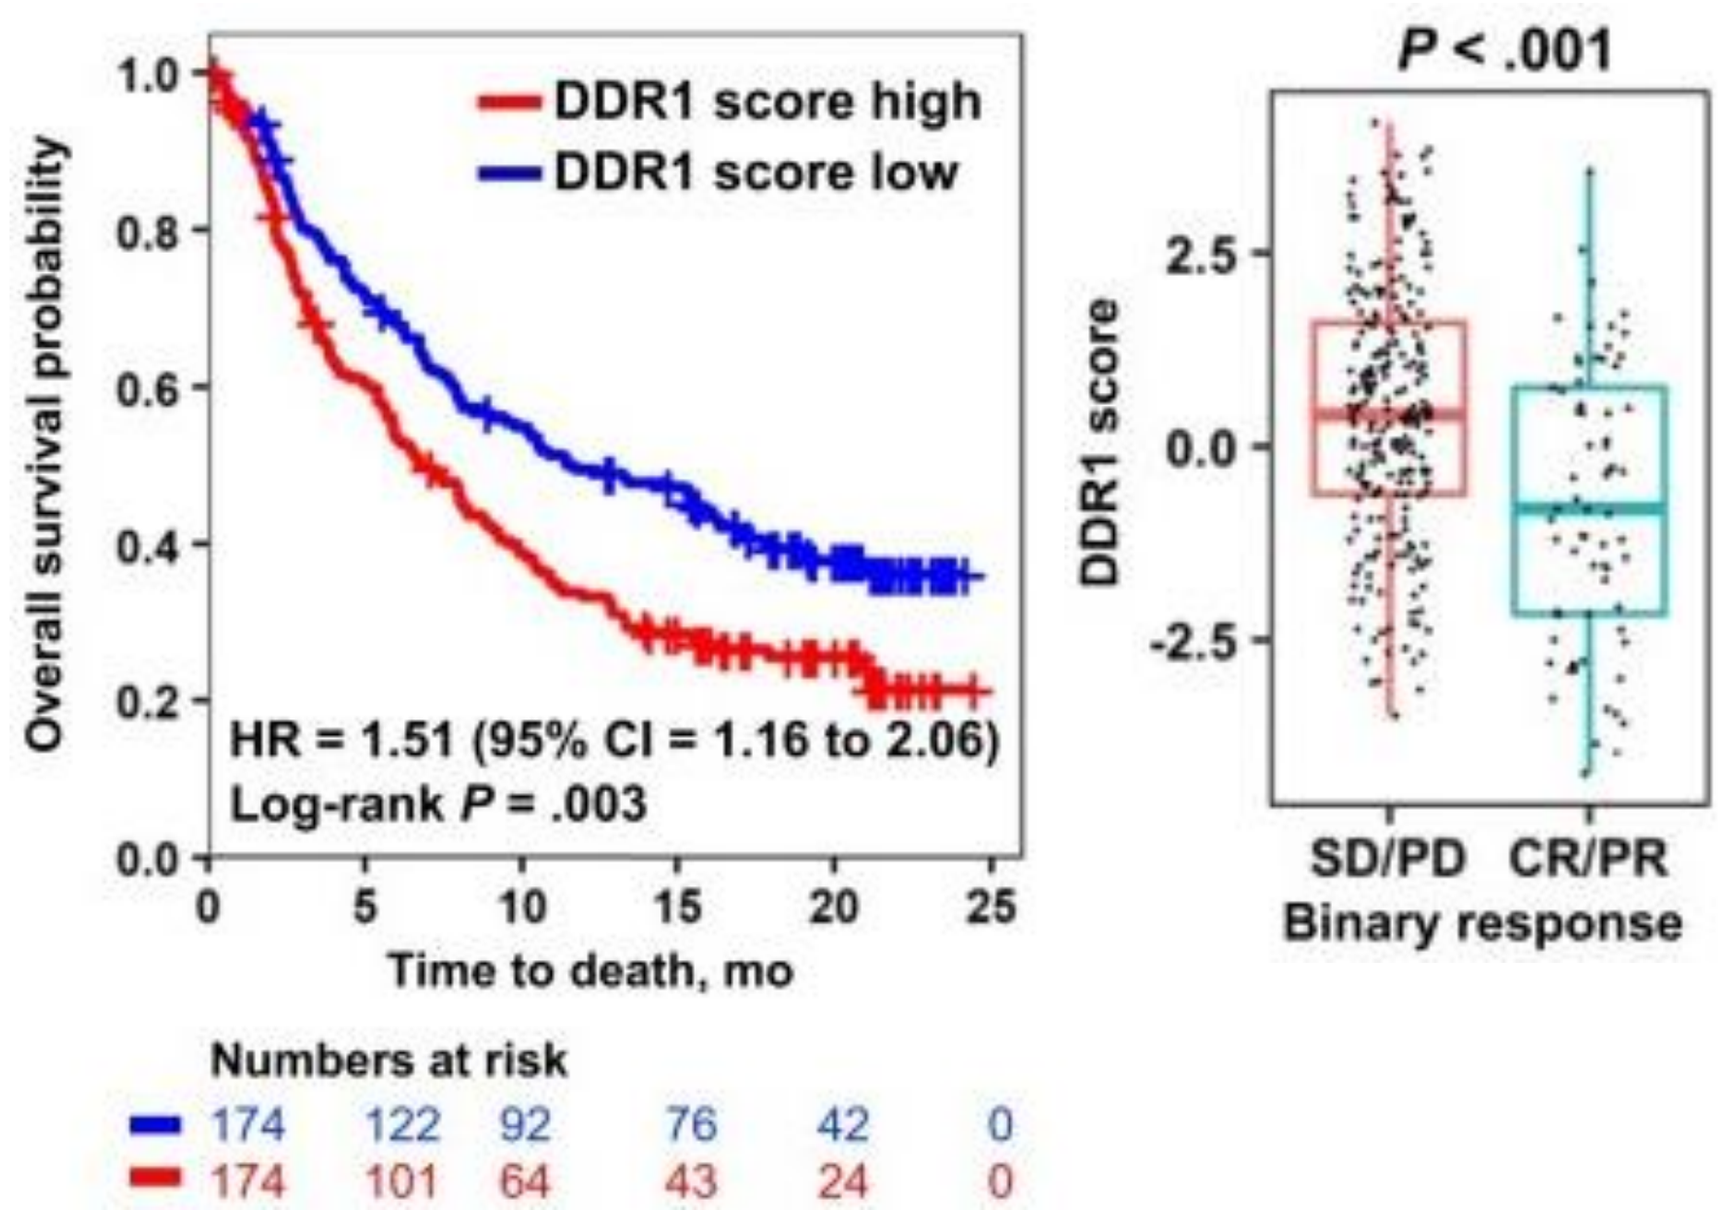
DDR1 score high
DDR1 score low
Overall survival probability
HR = 1.51 (95% CI = 1.16 to 2.06)
Log-rank P = .003
Time to death, mo
Numbers at risk
174 122 92 76 42 0
174 101 64 43 24 0
P < .001
DDR1 score
SD/PD
CR/PR
Binary response

- Secondary analyses of DDR1 expression using published data from the IMvigor210 trial (bladder cancer; NCT02108652) were conducted (You, *et al*. J Natl. Cancer Inst. 2022)
- Patients with high DDR1 scores who were treated with atezolizumab showed poor overall survival
- DDR1 scores were significantly higher in patients with stable disease or partial disease, compared to patients with complete response for partial response
- DDR1-high patients exhibited a non-T cell-inflamed phenotype
- The data suggest a high DDR1 score may predict lack of response to PD-1/PD-L1-targeted therapy

RESULTS

PRTH-101-001 Clinical Trial Design

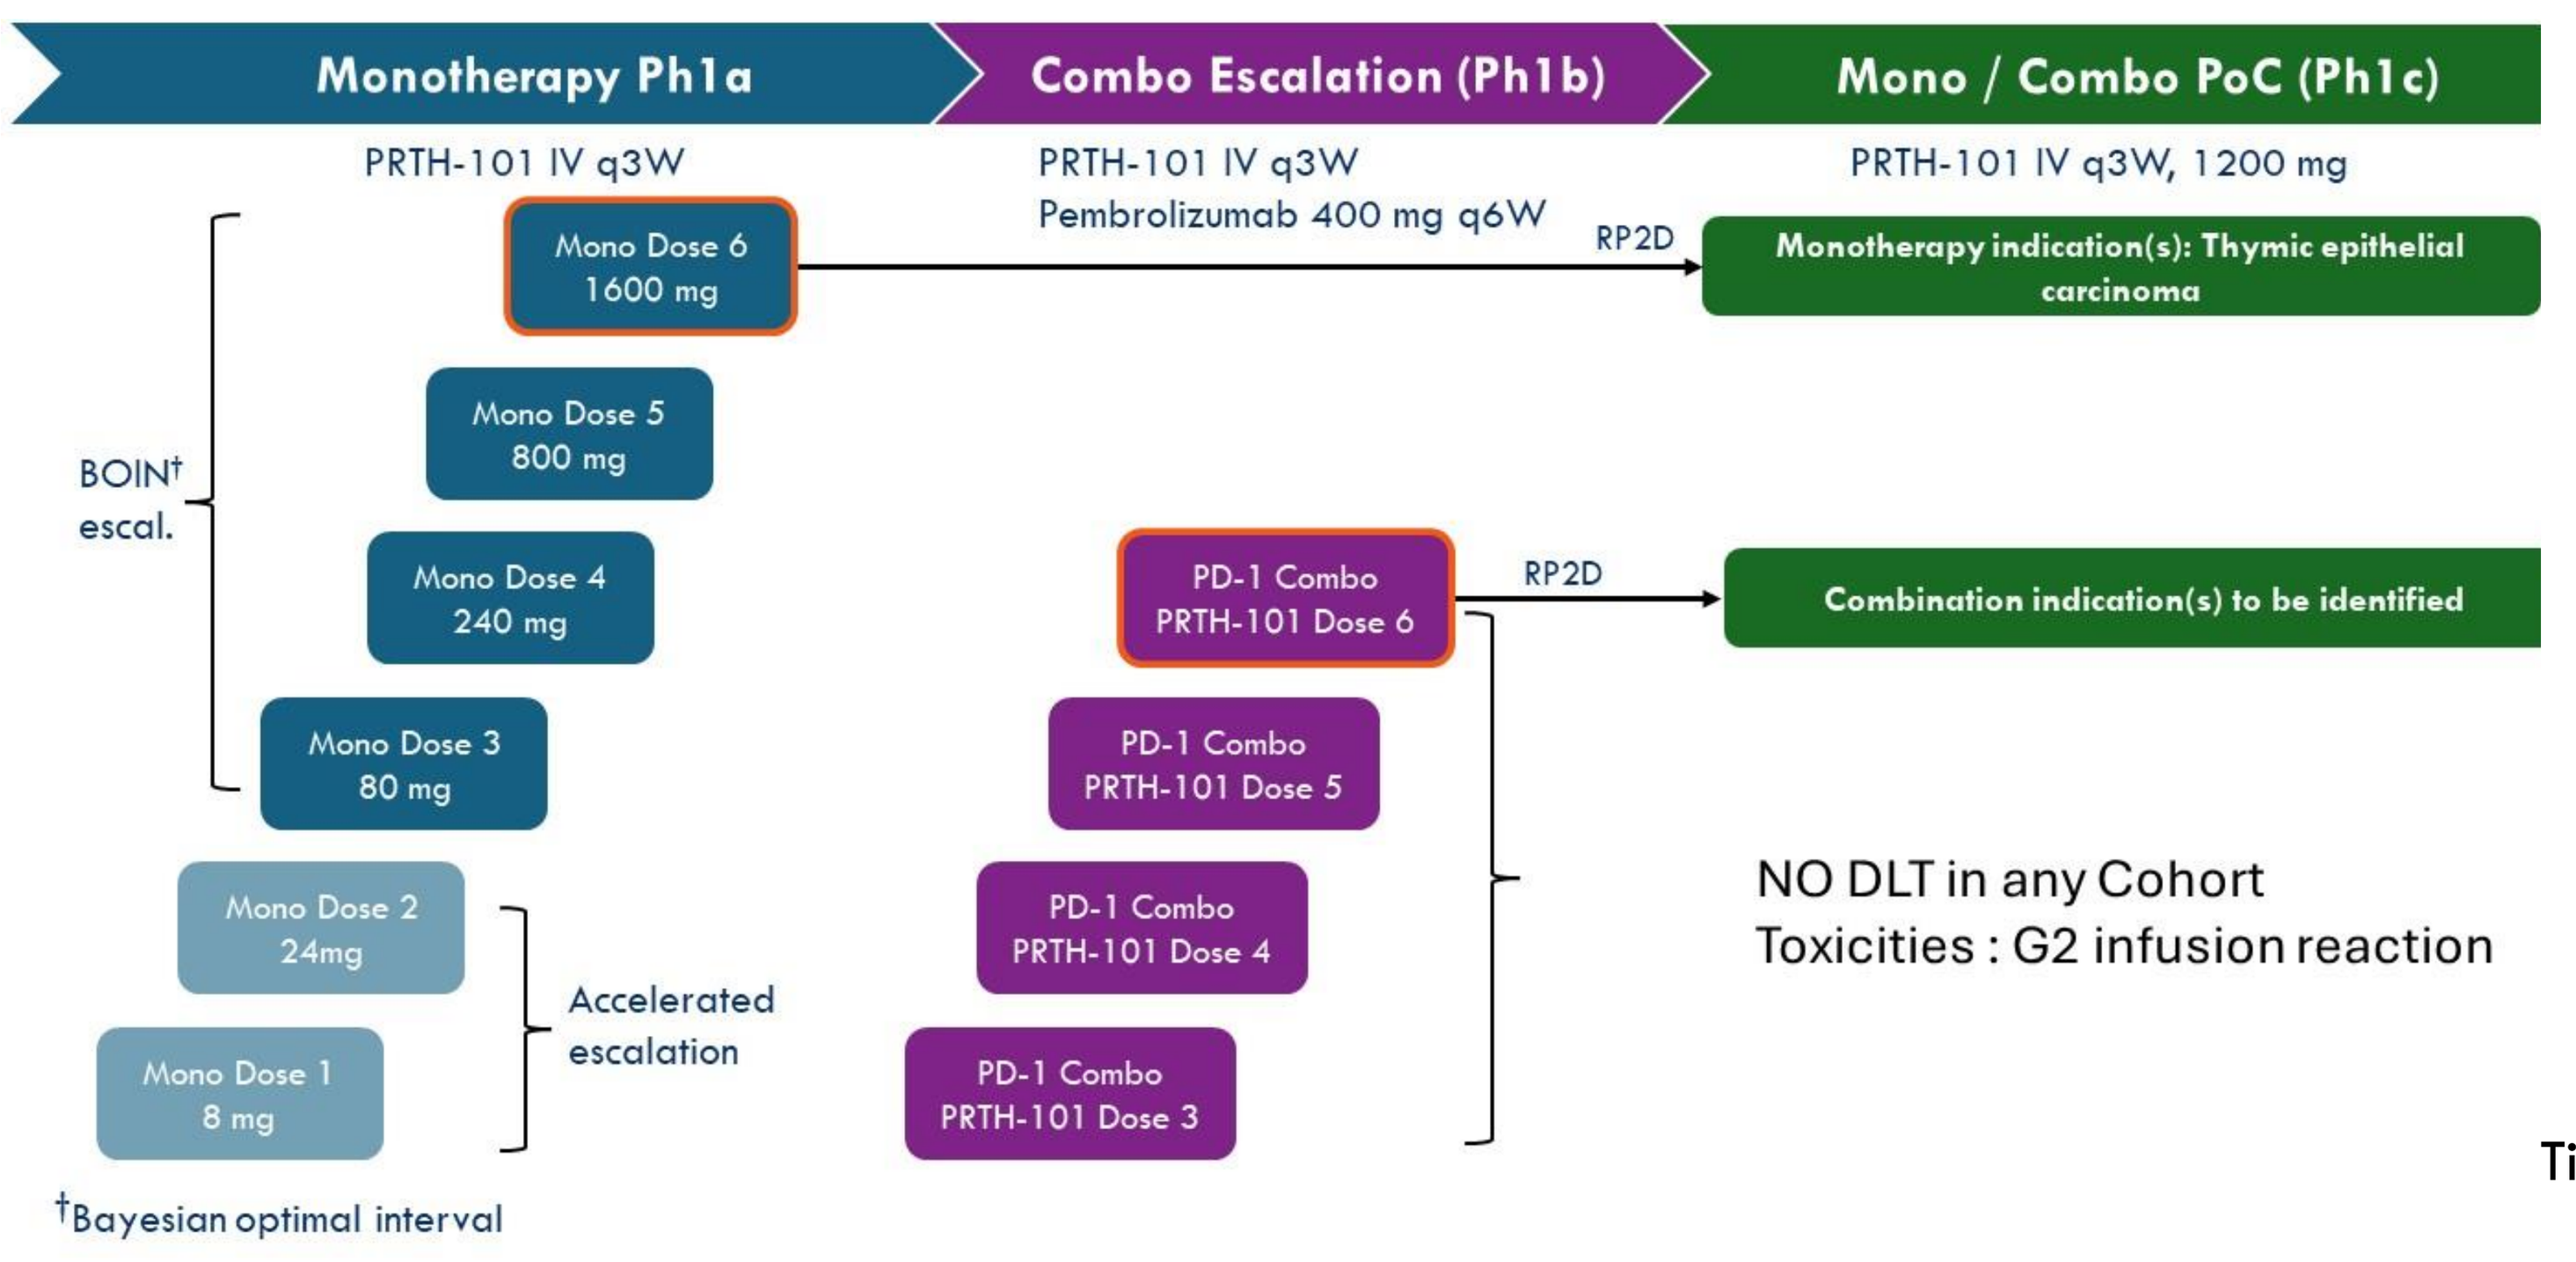
Monotherapy Ph1a
Combo Escalation (Ph1b)
Mono / Combo PoC (Ph1c)
PRTH-101 IV q3W
PRTH-101 IV q3W
Pembrolizumab 400 mg q6W
PRTH-101 IV q3W, 1200 mg
Mono Dose 6
1600 mg
RP2D
Monotherapy indication(s): Thymic epithelial carcinoma
Mono Dose 5
800 mg
BOIN† escal.
Mono Dose 4
240 mg
PD-1 Combo
PRTH-101 Dose 6
RP2D
Combination indication(s) to be identified
Mono Dose 3
80 mg
PD-1 Combo
PRTH-101 Dose 5
Mono Dose 2
24mg
PD-1 Combo
PRTH-101 Dose 4
Accelerated escalation
Mono Dose 1
8 mg
PD-1 Combo
PRTH-101 Dose 3
NO DLT in any Cohort
Toxicities : G2 infusion reaction
†Bayesian optimal interval

High DDR1 Expression in Thymic Tumors

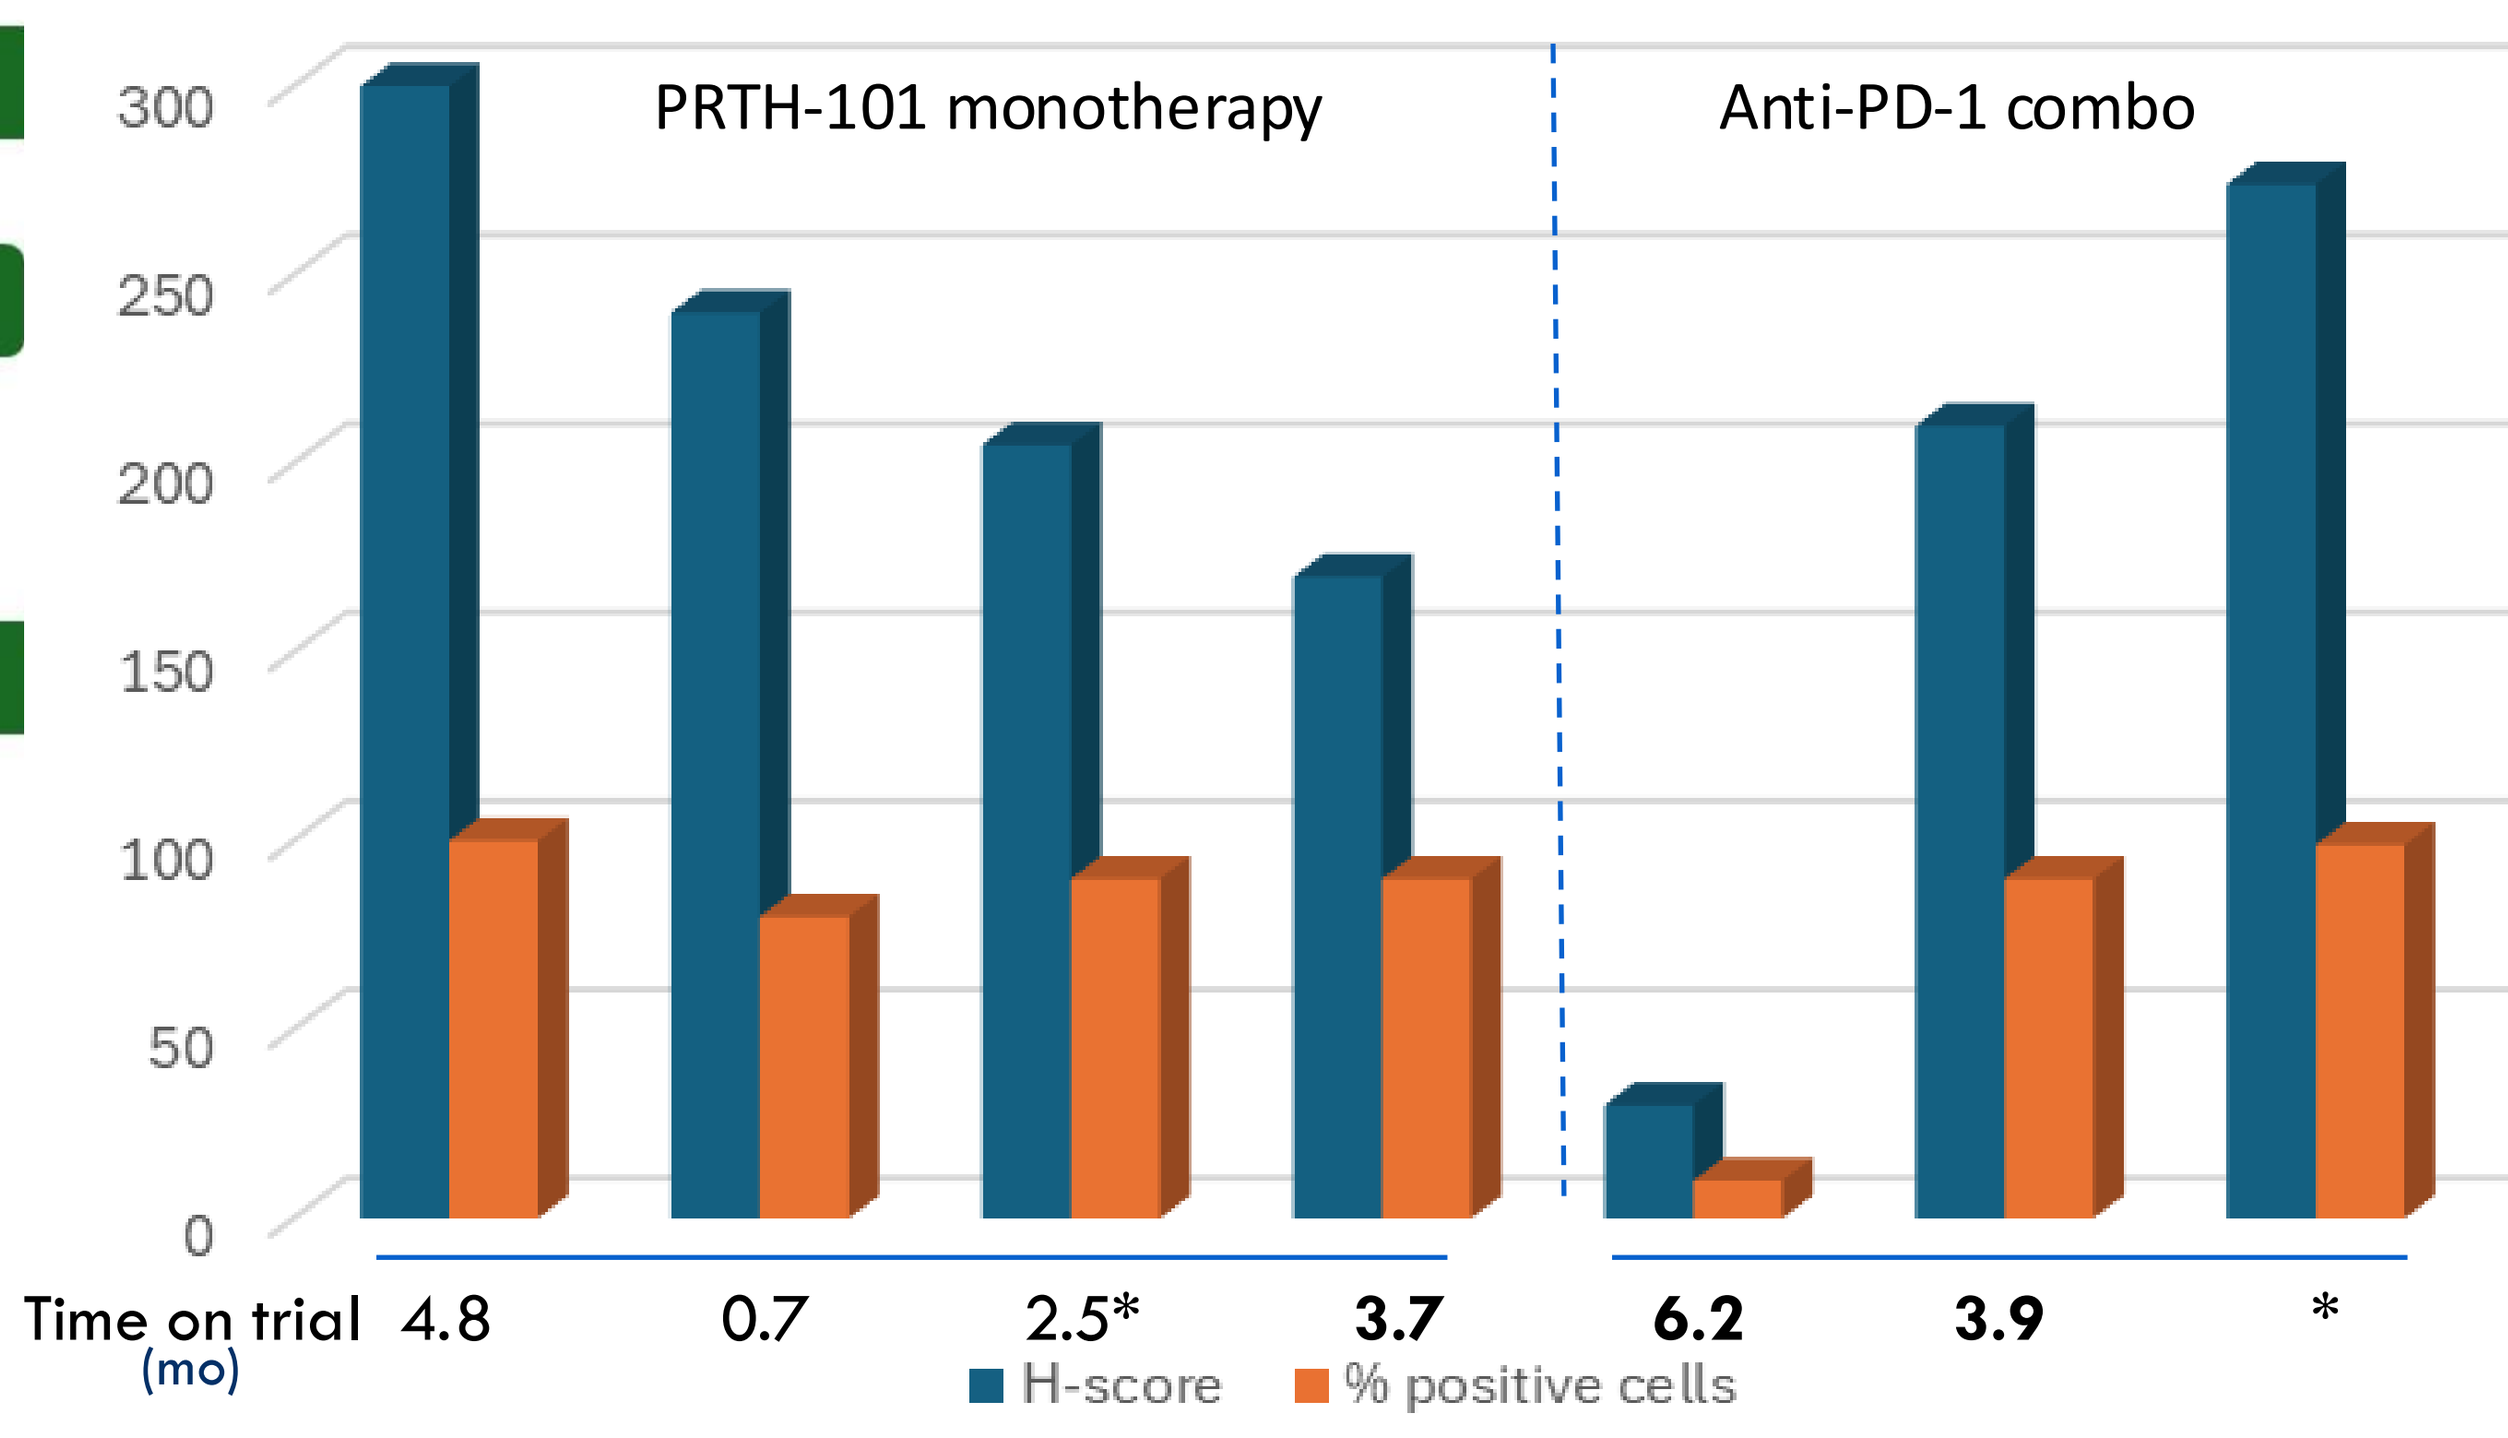
PRTH-101 monotherapy
Anti-PD-1 combo
Time on trial (mo)
4.8
0.7
2.5*
3.7
6.2
3.9
*
H-score
% positive cells

- All tumors expressed DDR1
- Overall, individual tumor cells expressed high levels of DDR1
- Baseline H-scores (histo score; 0 - 300) were calculated from slides stained for DDR1
- Scores were based on tumor membrane staining intensity (0, 1+, 2+, 3+) and percentage of cells at each

*discontinued due to unrelated AE; **Bold font** = ongoing

Study Summary (as of 1 October 2024)

- **Eligibility**
 - Metastatic or unresectable measurable solid malignancy for which no established therapy is of benefit, or has been declined by the patient
 - ECOG performance status 0 -1; no significant co-morbid illness
 - Prior PD-1/ PD-L1 therapy permissible; no prior ICI Adverse Events AE ≥ Grade 3 or > G2 myocarditis or pneumonitis. Immune-mediated AEs must have resolved to G1, except vitiligo, well-controlled endocrinopathies, or G2 peripheral neuropathy.
 - No uncontrolled CNS disease, hepatocellular cancer, sarcoma, or GBM
- **Status**
 - 45 subjects enrolled (enrollment complete for Phase 1a and Phase 1b); 11 ongoing
 - Median Age: 63 (range 25-83); Sex : M=28, F =17
 - Prior Therapies: median = 4 (range 1-8); prior immunotherapy = 17
 - Tumor Diagnoses:
 - CRC = 17; Thymic Carcinoma =10; PDAC = 6, NSCLC = 3; ovarian, SCCHN, thyroid = 2; bladder, SCLC, renal = 1
 - Time on study : median = 6 weeks (range < 3 to 63)
 - Dose limiting Toxicities = 0 (2 G2 infusion reactions)
 - Phase 1c dose determined (1200 mg); dosing initiated for thymic cohort
 - Projected completion of thymic cohort enrollment: Q1 2025

High DDR1 Expression is Significantly Associated with TGFβ Signaling in Thymic Tumors

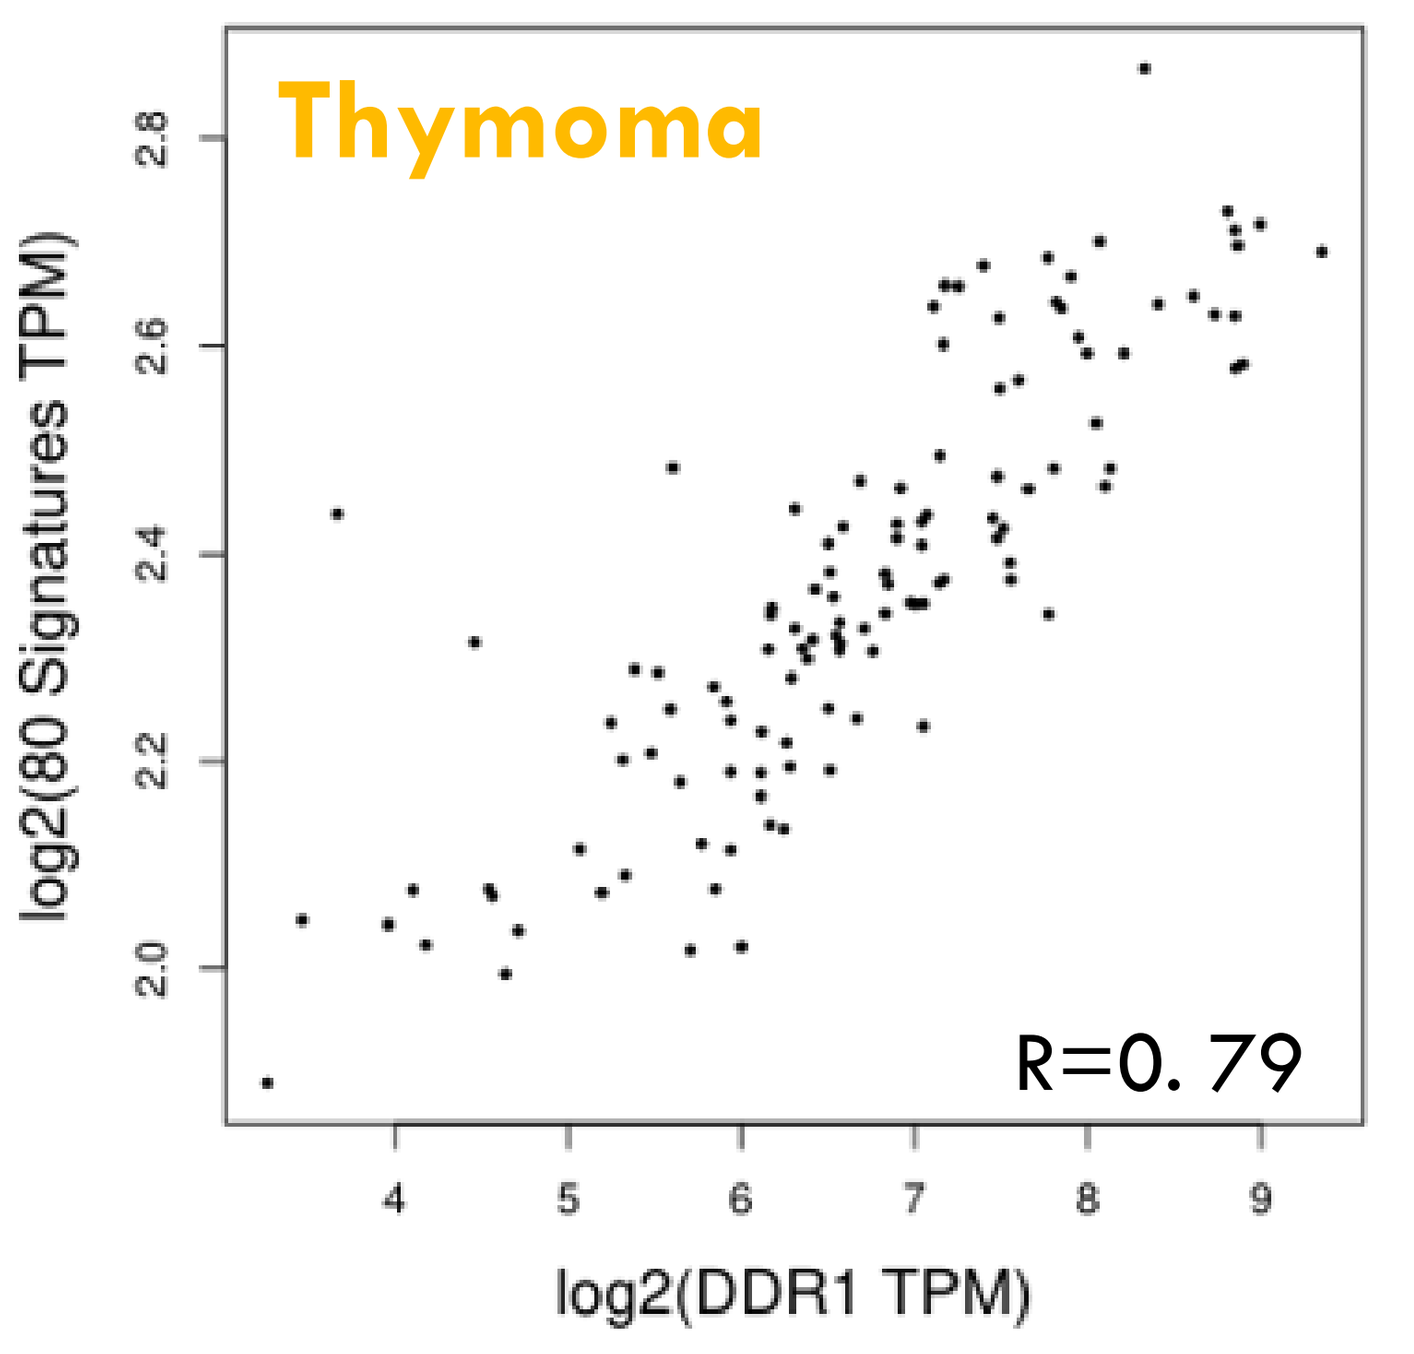
Thymoma
log2(80 Signatures TPM)
R=0. 79
log2(DDR1 TPM)

The Cancer Genome Atlas (TCGA) was queried for associations between an 80-gene TGFβ pathway activation signature and DDR1 gene expression.

$p<0.0015$ with Bonferroni correction

(Sher *et al*., SITC 2022.1461)

Status of Thymic Patients Treated with PRTH-101

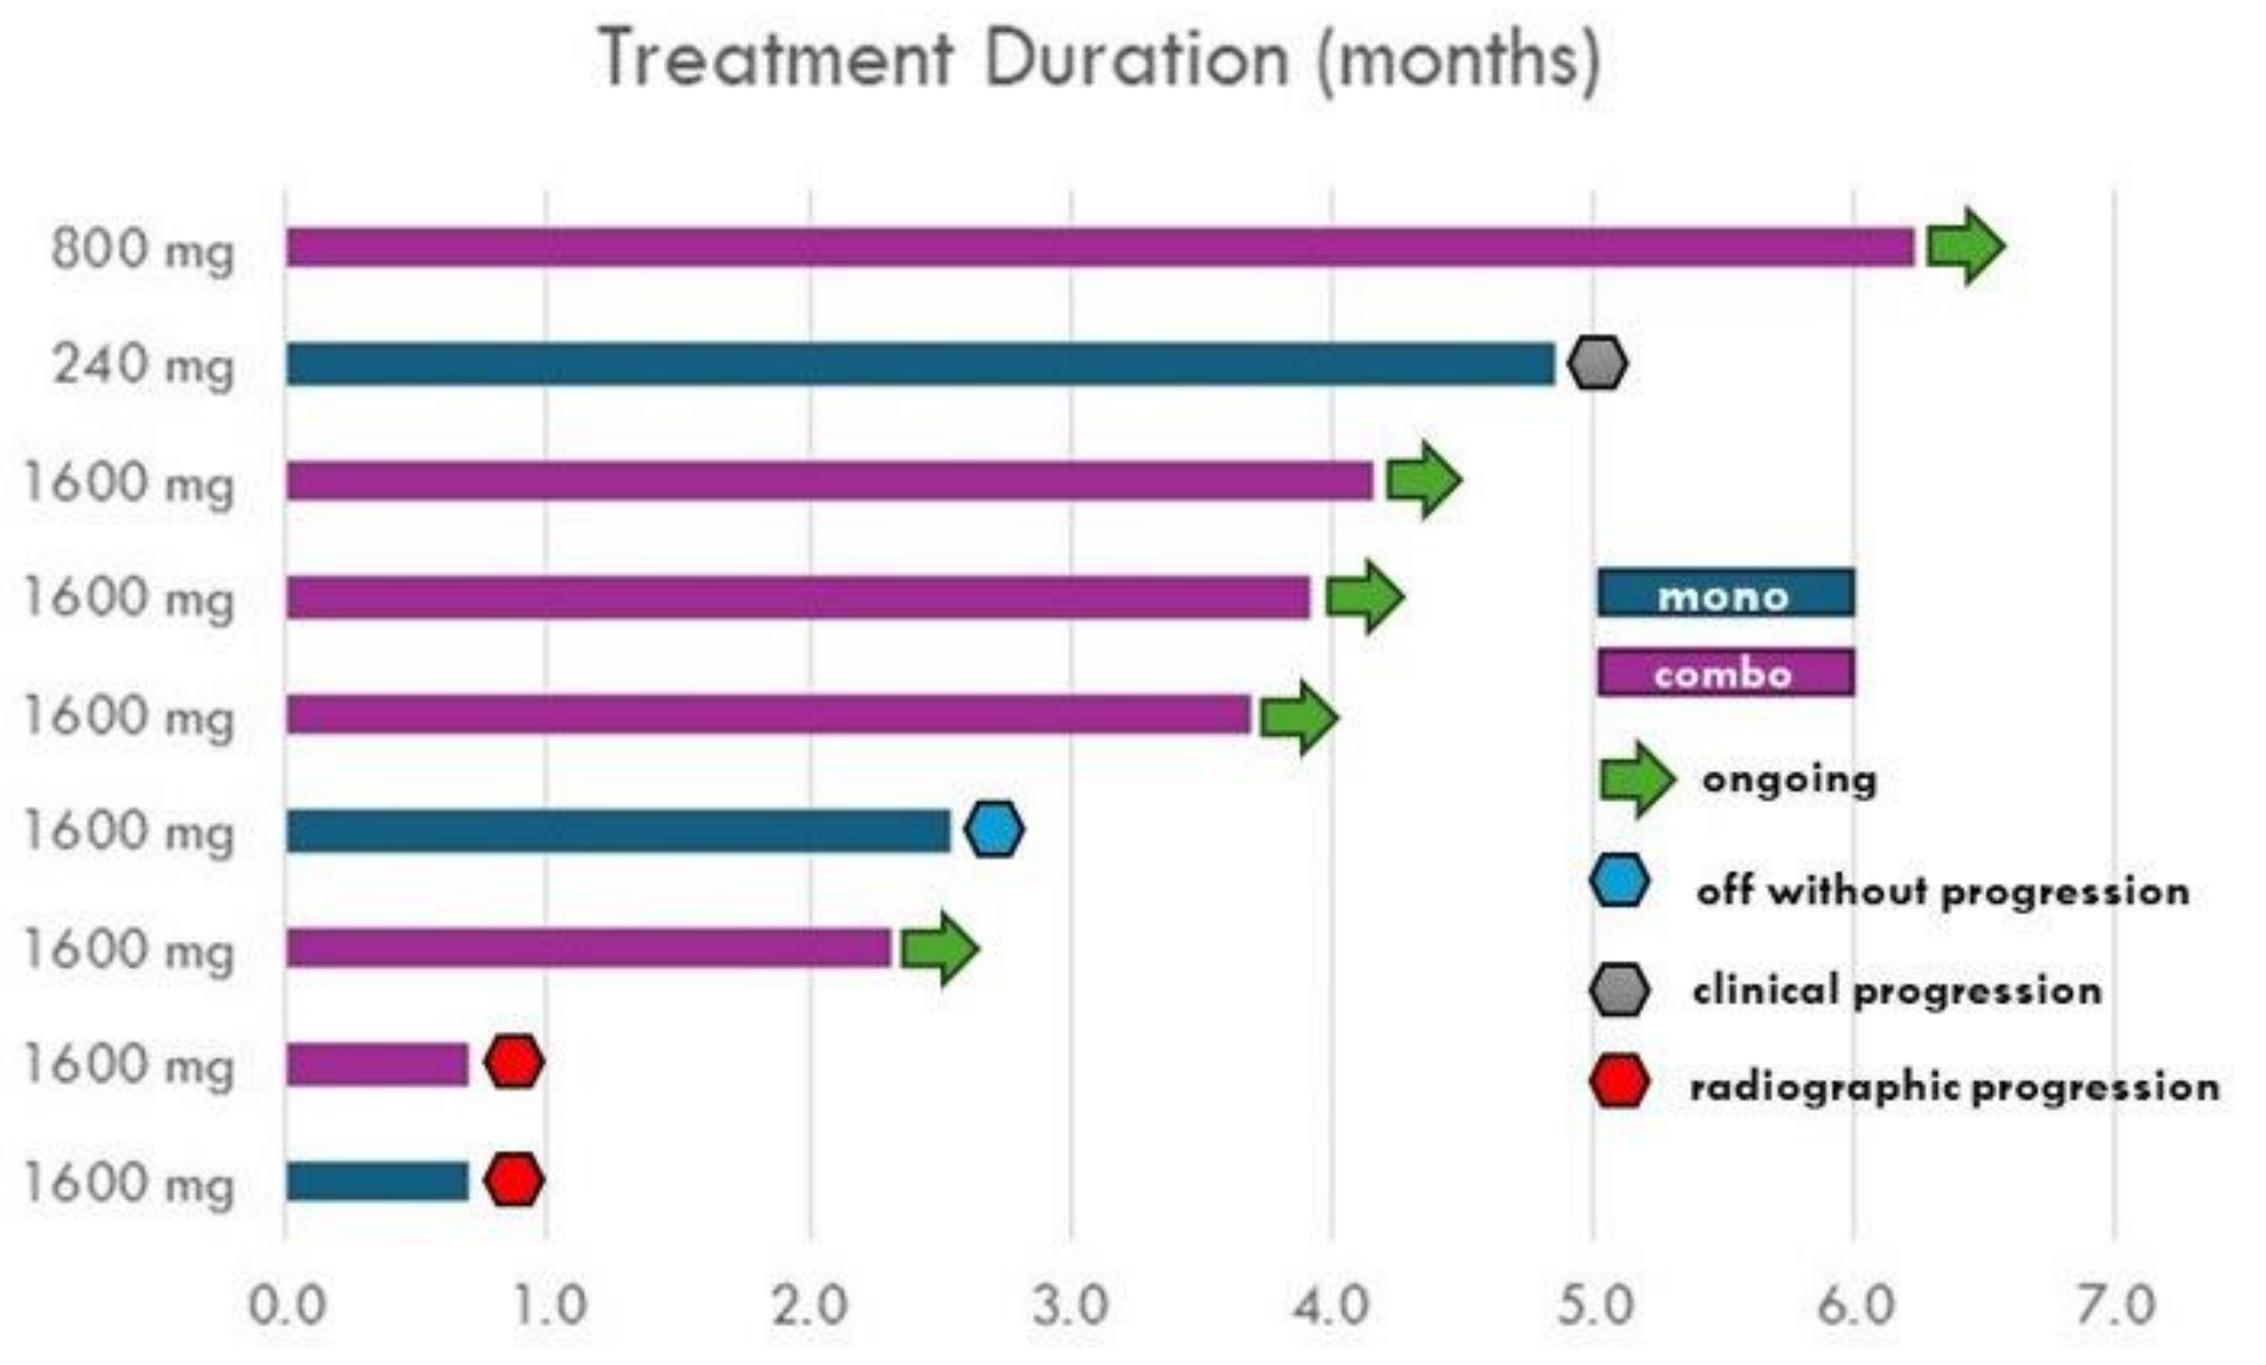
Treatment Duration (months)
800 mg
240 mg
1600 mg
1600 mg
1600 mg
1600 mg
1600 mg
1600 mg
1600 mg
mono
combo
ongoing
off without progression
clinical progression
radiographic progression

Reduction in Tumor Size Following Treatment with 1600 mg PRTH-101; Visual Improvement of Cutaneous Metastases

67-year-old male with metastatic thymic carcinoma (chest wall, lung, rib metastases)
Progression on prior treatments (carboplatin/paclitaxel, CX-072, AN4005, pemetrexed)

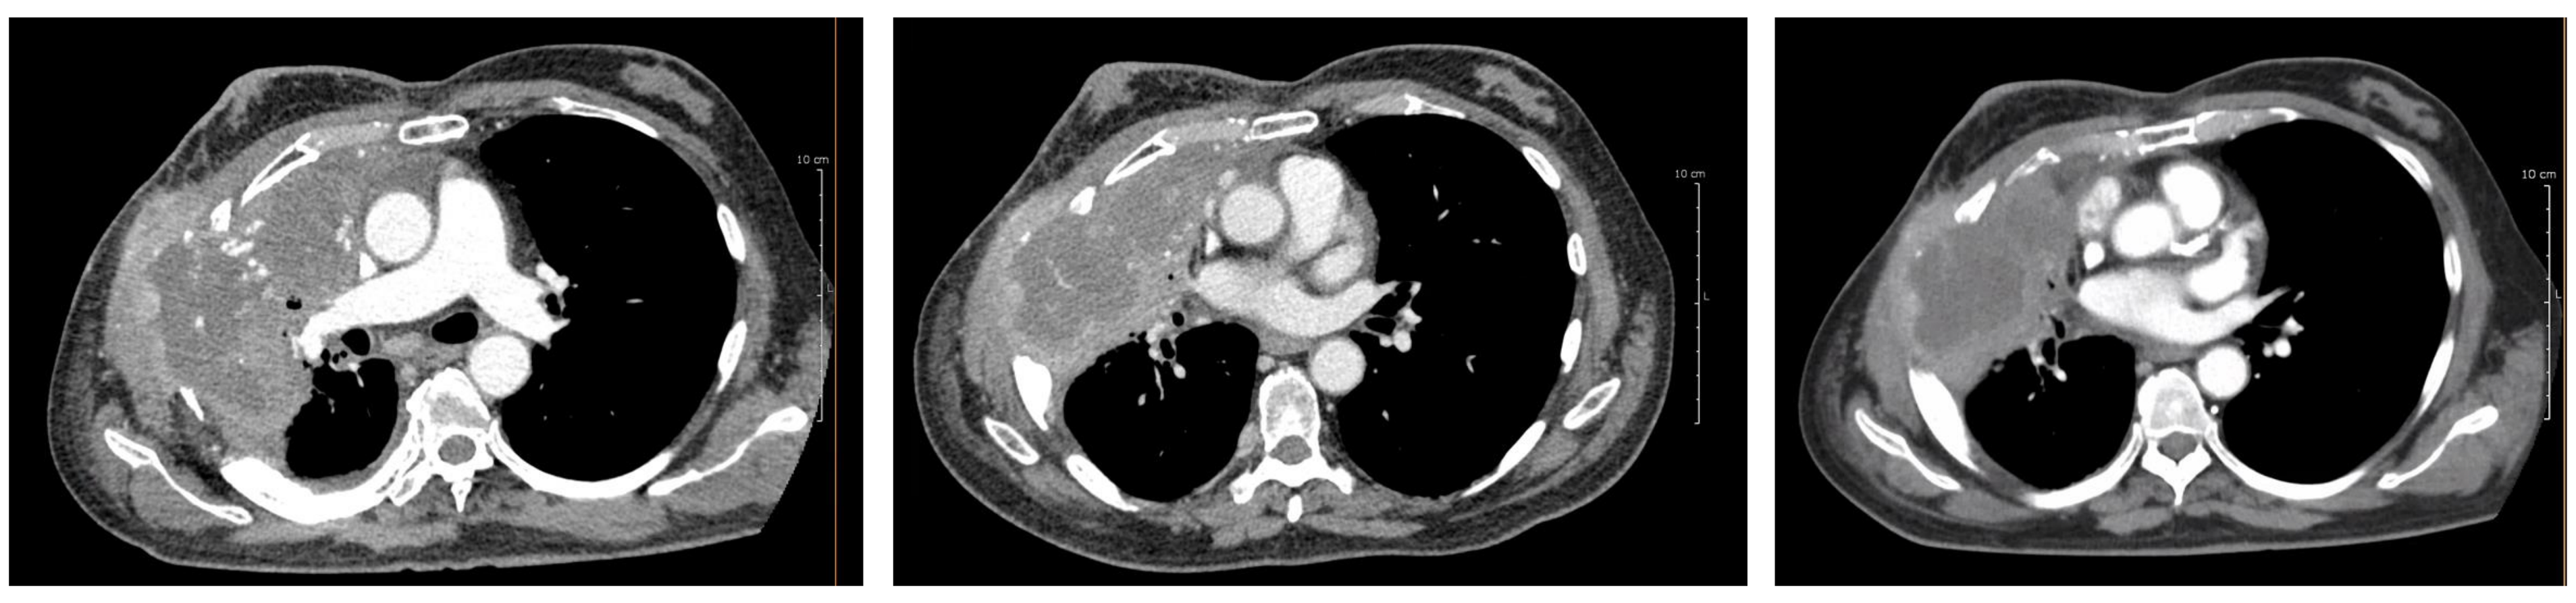

Screening | 6-wk scan | 12-wk scan

CONCLUSIONS

- **DDR1 is highly expressed in thymic cancers and other tumor types.**
- **DDR1 inhibition may facilitate tumor infiltration of T cells by disrupting collagen in the extracellular matrix of tumors and ameliorate clinical resistance to immune checkpoint inhibitors.**
- **PRTH-101 is a monoclonal antibody that blocks the interaction of collagen with DDR1 in the extracellular domain.**
- **PRTH-101 testing has shown no nonclinical toxicities or dose-limiting clinical toxicities in initial Phase 1 testing. The Phase 1c dose is 1200 mg.**
- **Phase 1 testing has shown early evidence of clinical benefit of PRTH-101 in thymic epithelial cancers.**

The Sponsor and Investigators are grateful to the patients and their families for their participation in this trial.

Drs. Eder, Schürpf, and Macdonald are employees of Incendia Therapeutics. Drs. Clifton and Dillon are former employees of Incendia Therapeutics. No other author has any affiliation with Incendia Therapeutics.

INCENDIA THERAPEUTICS